Poster no-1335

In-vitro activity of solithromycin against anaerobic bacteria in the normal intestinal microbiota

A. WEINTRAUB - M. RASHID - C.E. NORD

Division of Clinical Microbiology, Department of Laboratory Medicine, Karolinska University Hospital, Karolinska Institutet, Stockholm, Sweden

Introduction and Purpose:

The normal microbiota acts as a barrier against colonization by potentially pathogenic microorganisms and against overgrowth of already present opportunistic microorganisms. Administration of antimicrobial agents, therapeutically or as prophylaxis, causes disturbances in the ecological balance between the host and the normal microbiota. Solithromycin is a fourth generation macrolide and first fluoroketolide. It is being developed as intravenous and oral formulations for the treatment of patients with community-acquired bacterial pneumonia (CABP). Solithromycin inhibits bacterial protein synthesis via a unique ribosomal binding pattern to domains II and V, as well as to the peptide tunnel, of the 23S component of the 50S ribosomal subunit (1). The objective was to determine the *in-vitro* activity of solithromycin in comparison to amoxicillin/clavulanic acid, azithromycin, ceftriaxone, metronidazole and levofloxacin by determining the minimum inhibitory concentration (MIC) against the normal anaerobic intestinal microbiota.

Methods:

1024 anaerobic strains including bifidobacteria, lactobacilli, clostridia, bacteroides, prevotella and veillonella were isolated from the faecal samples of healthy volunteers that had no history of systemic antibiotics within the past 3 months and no medical conditions that can alter their health or microbiota. Faecal samples were diluted from tenfold to 10^7 and inoculated on non-selective and selective agars. The agar plates were incubated for 2-7 days at 37°C in anaerobic jars. After incubation, different colony types were analysed according to Gram-reaction and colony morphology, and identified by MALDI-TOF. In addition, 28 *Clostridium difficile* isolates from patients with a primary *C. difficile* infection at the Karolinska University Hospital were investigated.

Table 1. MIC values of 1024 anaerobic bacteria from the normal human microbiota and 28 clinical isolates of *C. difficile* against 6 different antimicrobial agents

Bacteria (No. of strains)	Antimicrobial agent mg/l								
	Solithromycin			Amoxicillin-clavulanic acid			Azithromycin		
	MIC_{50}	MIC_{90}	Range	MIC_{50}	MIC_{90}	Range	MIC_{50}	MIC_{90}	Range
Bifidobacteria (254)	0.008	0.008	0.008-0.5	0.032	0.25	0.008-0.25	0.016	0.25	0.008-256
Lactobacilli (257)	0.008	0.016	0.008-0.5	0.125	0.5	0.008-1.0	0.25	256	0.008-256
Clostridia (225)	0.5	8.0	0.008-128	0.25	32	0.008-256	128	256	0.032-256
Bacteroides (255)	0.5	4.0	0.032-256	0.5	4.0	0.016-64	32	256	0.5-256
Prevotella (26)	0.125	2.0	0.016-4.0	0.25	0.5	0.125-1.0	1.0	128	0.5-128
Veillonella (7)	0.016	0.032	0.016-1.0	0.25	0.25	0.25-0.25	0.5	4.0	0.5-32
C. difficile (28)	0.032	0.064	0.032-4.0	0,5	0,5	0.25-0.5	0.008	0.5	0.008-4.0

Bacteria (No. of strains)	Antimicrobial agent mg/l								
	Ceftriaxone			Metronidazole			Levofloxacin		
	MIC_{50}	MIC_{90}	Range	MIC_{50}	MIC_{90}	Range	MIC_{50}	MIC_{90}	Range
Bifidobacteria (254)	0.125	4.0	0.008-16	4.0	256	0.008-256	2.0	8.0	0.008-256
Lactobacilli (257)	32	256	0.008-256	256	256	256-256	2.0	32	0.008-128
Clostridia (225)	32	256	0.032-256	0.5	64	0.032-256	4.0	8.0	0.25-32
Bacteroides (255)	32	256	0.125-256	0.5	32	0.016-256	4.0	16	0.032-64
Prevotella (26)	8	128	1.0-128	0.5	1.0	0.032-1.0	4.0	8.0	0.016-8.0
Veillonella (7)	16	16	16-16	256	256	256-256	2.0	2.0	2.0-4.0
C. difficile (28)	16	16	16-32	0.064	0.064	0.064-0.125	2.0	2.0	2.0-2.0

The MICs of solithromycin, amoxicillin/clavulanic acid, azithromycin, ceftriaxone, metronidazole and levofloxacin were determined by the agar dilution method. The final inoculum was 10^5 colony forming units (CFU) per spot. Inoculated plates were incubated for 48 h at 37°C in anaerobic jars. Reference strains were *Bacteroides fragilis* ATCC 25285, *Clostridium difficile* ATCC 700057 and *Eubacterium lentum* ATCC 43055 (2).

Results:

The MICs for solithromycin against bifidobacteria (254) were: MIC_{50}, 0.008 mg/l; MIC_{90}, 0.008 mg/l; MIC range, 0.008-0.5 mg/l; lactobacilli (257): MIC_{50}, 0.008 mg/l; MIC_{90}, 0.016 mg/l; MIC range, 0.008-0.5 mg/l; clostridia (225): MIC_{50}, 0.5 mg/l; MIC_{90}, 8.0 mg/l; MIC range, 0.008-128 mg/l; bacteroides (255): MIC_{50}, 0.5 mg/l; MIC_{90}, 4.0 mg/l; MIC range, 0.032-256 mg/l; prevotella (26): MIC_{50}, 0.125 mg/l; MIC_{90}, 2.0 mg/l; MIC range, 0.016-4.0 mg/l; veillonella (7): MIC_{50}, 0.016 mg/l; MIC_{90}, 0.032 mg/l; MIC range, 0.016-1.0 mg/l; *C. difficile* (28): MIC_{50}, 0.032 mg/l; MIC_{90}, 0.064 mg/l; MIC range, 0.032-4.0 mg/l. Most of the Gram-positive anaerobic bacteria were sensitive to amoxicillin-clavulanic acid but less sensitive to azithromycin, ceftriaxone and levofloxacin. Bifidobacteria and lactobacilli were resistant to metronidazole.

Bacteroides strains showed high MIC-values for azithromycin, ceftriaxone and levofloxacin and low MIC-values for amoxicillin-clavulanic acid, metronidazole. Similar MIC patterns were observed in *Prevotella*. *Veillonella* strains showed high MIC values for metronidazole and ceftriaxone and lower values for amoxicillin-clavulanic acid and azithromycin.

Conclusions:

Solithromycin is active against bifidobacteria, lactobacilli and most clostridia isolated from the normal intestinal microbiota and most of the *C. difficile* clinical isolates. Solithromycin is less active against bacteroides strains which may explain the protective role in developing *C. difficile* infections (3).

References:

1. Llano-Sotelo B, Dunkle J, Klepacki D, Zhang W, Fernandes P, Cate JH, et al 2010. Binding and action of CEM-101, a new fluoroketolide antibiotic that inhibits protein synthesis. Antimicrob Agents Chemother 54:4961-70
2. CLSI. Methods for antimicrobial susceptibility testing of anaerobic bacteria; Approved standard. 8th ed. Document M11-A8. Wayne, PA: CLSI; 2012.
3. Louie TJ, Emery J, Krulicki W, Byrne B, Mah M. 2009. OPT-80 eliminates *Clostridium difficile* and is sparing of bacteroides species during treatment of *C. difficile* infection. Antimicrob Agents Chemother 53:261-263.

Acknowledgements:

The study was supported by a grant from Cempra Inc, Chapel Hill, NC, USA. The skilful technical assistance of Monica Sörensson is gratefully acknowledged.

Corresponding author:
Carl Erik Nord, Professor
Telephone: +46 8 585 87838
Fax: +46 8 585 87933
E-mail address: carl.erik.nord@ki.se