Antibiotic Treatment Patterns And Outcomes Among Patients Hospitalised in ICU With Community-Acquired Bacterial Pneumonia: A US Cohort Study

Sara Eapen, PhD[1]; Edward G. Tuttle, MBA[1]; Christopher J. Llop, BSc[1]; Matthew Barrett, PhD[1]

[1] Analysis Group, Inc., Boston, MA

Programme Number: eP066

INTRODUCTION

- Community-acquired bacterial pneumonia (CABP) has an annual incidence rate of 5–11 per 1,000 population worldwide and is associated with considerable morbidity and mortality, with up to 68.8% of patients requiring hospitalization[1–5]
- Rapid selection and initiation of appropriate antibiotic therapy is vital, shortening the illness course and significantly reducing the risk of complications or mortality[6]
- However, treatment decisions are complicated by the difficulty of obtaining timely microbiological diagnosis, antibiotic resistance and the numerous choices of generic and branded therapy available
- The purpose of this study was to review current clinical practices in the US and assess the relationship of antibiotic use in CAP patients hospitalised in the ICU [or CCU] on the following outcome measures:
 - In-hospital mortality
 - Rate of CABP related rehospitalisations at 30 days post index CABP episode
 - In-hospital rate of adverse events
 - Length of hospital stay

MATERIALS AND METHODS

Data Source:

- This retrospective cohort observational study was conducted using the Truven Health MarketScan Hospital Drug Database (HDD)
 - Covering January 2007 through December 2012
 - Including data from about 600 US-based hospitals (teaching and non-teaching)
 - Capturing de-identified in-patient information on demographic and clinical characteristics, medical and pharmacy resource utilization, as well as hospitalization costs

Study Population:

- Inclusion criteria:
 - Patients with primary diagnosis of CABP (ICD-9-CM: 481.xx, 482.xx, 484.xx–486.xx)
 - Patients treated with antibiotic therapy within 48 hours of hospital admission
 - Patients admitted to the ICU [or CCU] within the first 24 hours of admission
 - Patients ≥18 years of age
- Exclusion criteria:
 - Patients with tuberculosis (010.xx–018.xx, 647.33, 647.34, 647.30, 647.30, 647.32), cystic fibrosis (277.xx), HIV (042.xx), other pneumonia (055.1x, 073.0x, 115.05, 115.15, 115.95, 003.22, 480.xx, 487.0, 488.01, 488.11, 517.1, 770.0, 997.31, 488.81, 997.32), acute respiratory failure (518.81), septicemia NOS (038.9x), septic shock (785.52), severe sepsis (995.92), or acute renal failure NOS (584.9x)
- Index hospitalization was defined as the patient's initial CABP hospitalization observed in the data

Outcome Measures:

- Patient demographic and clinical characteristics (age, gender, comorbidities)
- Description of initial antibiotic therapy
- In-hospital mortality
- Rate of CABP related rehospitalisations at 30 days post index CABP episode
- In-hospital rate of adverse events. Adverse events evaluated included:
 - *Clostridium difficile* and enterocolitis
 - Peripheral neuropathy
 - Events related to the central nervous system, skin, gastrointestinal system
 - Hepatotoxicity
 - Hematologic toxicity
 - QTc interval prolongation
- Length of hospital stay

Statistical Analysis:

- Frequency counts and percentages were used to summarize categorical variables; means and standard deviations were reported for continuous variables
- Categorical outcome variables were compared, by type of initial antibiotic therapy, using Chi-Squared test and Wilcoxon Ranked-Sum test was used for continuous outcome variables
- Rates of adverse events, mortality and rehospitalisation were compared, by type of initial antibiotic therapy, using multivariate models that controlled for age, gender, comorbidities, and length of hospital stay

REFERENCES

1. Woodhead M: Community-acquired pneumonia in Europe: causative pathogens and resistance patterns. *Eur Respir J Suppl* 2002, 36:20s–27s.
2. Welte T: Risk factors and severity scores in hospitalized patients with community-acquired pneumonia: prediction of severity and mortality. *Eur J Clin Microbiol Infect Dis* 2012, 31:33–47.
3. Welte T, Kohnlein T: Global and local epidemiology of community-acquired pneumonia: the experience of the CAPNETZ Network. *Semin Respir Crit Care Med* 2009, 30:127–135.
4. http://www.medscape.com/viewarticle/737813_2. Accessed April 29, 2014
5. Capelastegui A, Espana PP, Bilbao A, Gamazo J, Medel F, Salgado J, Gorostiaga I, Esteban C, Altube L, Gorordo I, Quintana JM: Study of community-acquired pneumonia: incidence, patterns of care, and outcomes in primary and hospital care. *J Infect* 2010, 61:364–371.
6. Woodhead M, Blasi F, Ewig S, Garau J, Huchon G, Ieven M, Ortqvist A, Schaberg T, Torres A, van der Heijden G, Read R, Verheij TJ: Guidelines for the management of adult lower respiratory tract infections--full version. *Clin Microbiol Infect* 2011, 17(Suppl 6):E1–E59.
7. http://www.med.unc.edu/medselect/files/Guidelinesforpneumoniatreatment.pdf. Accessed April 9, 2014.

RESULTS

- There were a total of 60,548 patients admitted to the ICU with a primary diagnosis of CABP and 38,449 received treatment within 48 hours of admission
- 30,284 (50%) met all pre-specified selection criteria and were included in the study; 41% of whom (N=12,413) were treated according to guidelines*
- The guidelines-indicated cohort had 4,390 fluoroquinolone + beta lactam (F/BL) and 8,023 macrolide + beta lactam (M/BL) patients
 - The most common regimens within the two groups were levofloxacin + ceftriaxone sodium [N=1,688] and azithromycin + ceftriaxone sodium [N=6,836], respectively
- Figure 1 depicts the patient selection process

Figure 1: Patient selection flow chart

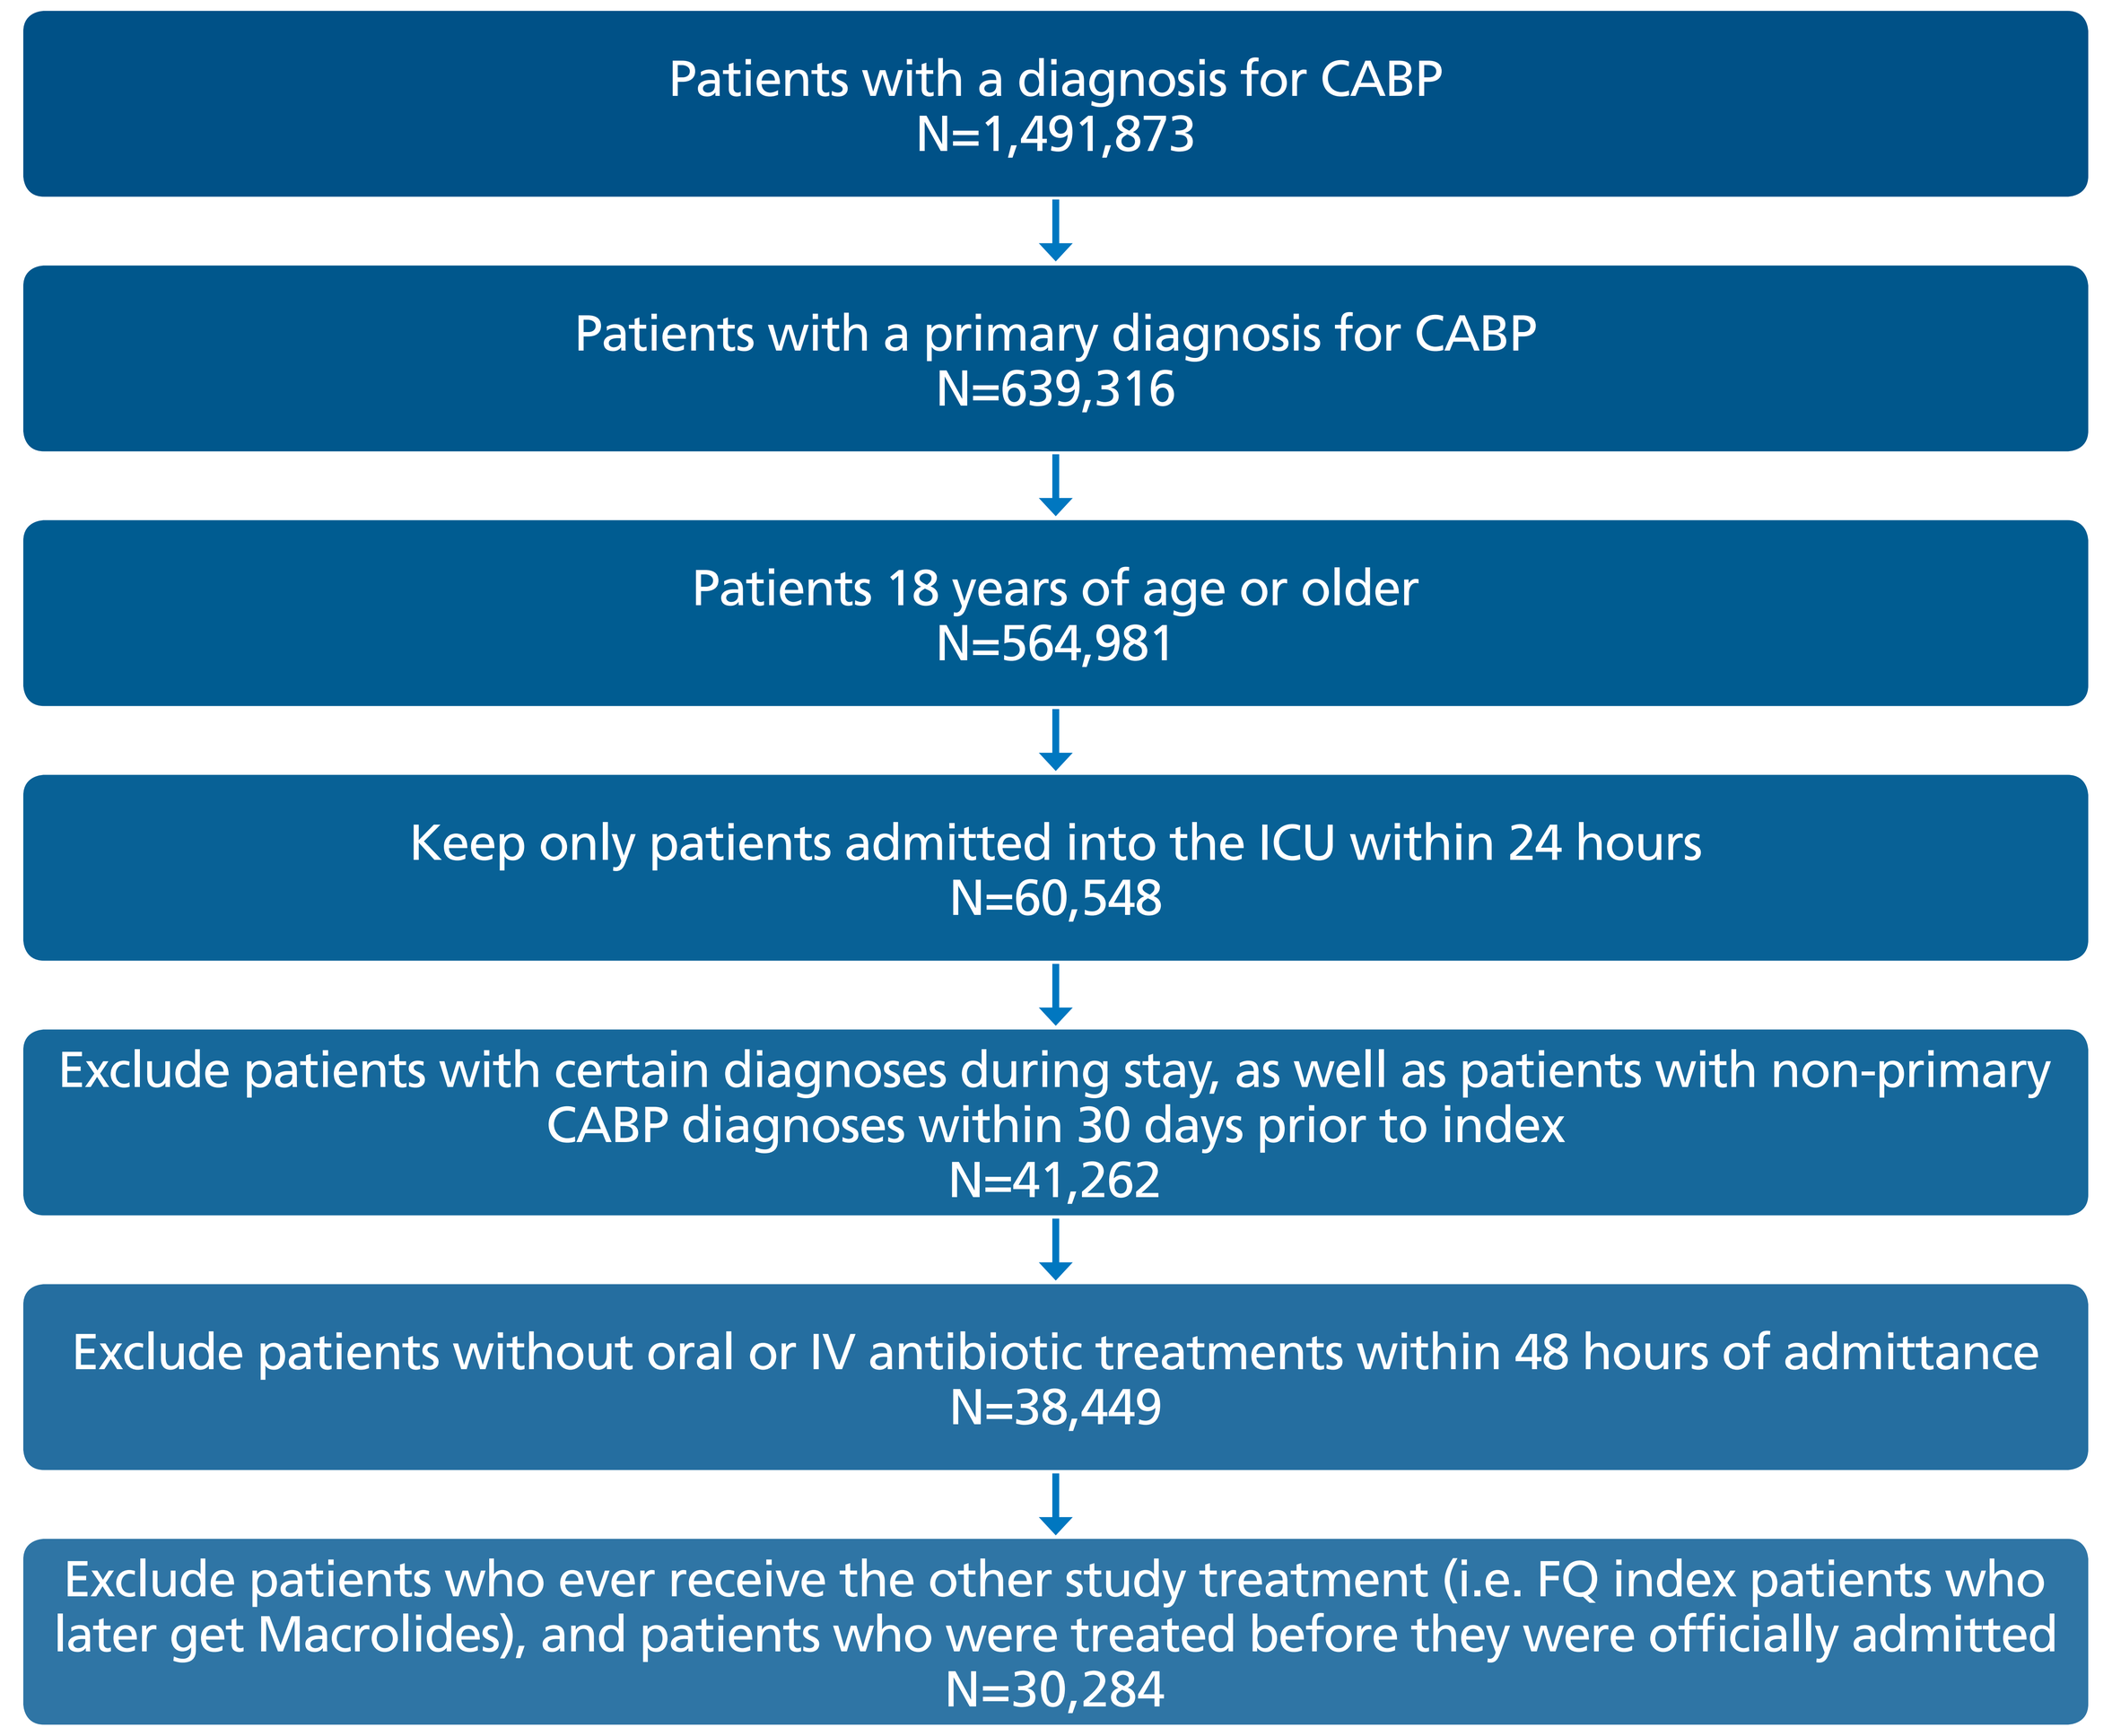
Patients with a diagnosis for CABP
N=1,491,873
Patients with a primary diagnosis for CABP
N=639,316
Patients 18 years of age or older
N=564,981
Keep only patients admitted into the ICU within 24 hours
N=60,548
Exclude patients with certain diagnoses during stay, as well as patients with non-primary CABP diagnoses within 30 days prior to index
N=41,262
Exclude patients without oral or IV antibiotic treatments within 48 hours of admittance
N=38,449
Exclude patients who ever receive the other study treatment (i.e. FQ index patients who later get Macrolides), and patients who were treated before they were officially admitted
N=30,284

* For patients in the ICU setting or with severe CABP, ATS, IDSA, ERS and BTS guidelines recommend BL (cefotaxime, ceftriaxone, or ampicillin-sulbactam) plus either azithromycin or a fluoroquinolone.

- Patients receiving fluoroquinolone regimens were slightly older and had more comorbidities relative to the M/BL patients at index hospitalisation

Table 1: Key patient demographic and clinical characteristics*

Parameters	Fluoroquinolone + Beta-Lactam N=4,390	Macrolide + Beta-Lactam N=8,023
Age (yrs), Mean ± SD	70.1 ± 16.0	68.5 ± 16.9
Total Length of Stay (days), Mean ± SD	5.5 ± 4.3	4.5 ± 3.2
Health Index**, Mean ± SD	2.2 ± 1.9	2.0 ± 1.7
Male, %	49.8%	49.2%

* P-value<0.05 for Age, Length of Stay, and Health Index.

** Health Index was calculated by evaluating the presence of the following diagnoses during index hospitalisation: Myocardial Infarction (410.xx, 412.xx), Congestive Heart Failure (402.01, 402.11, 402.91, 404.01, 404.03, 404.11, 404.13, 404.91, 404.93, 425.xx, 428.xx, 429.3x), Peripheral Vascular Disease (440.xx, 441.xx, 442.xx, 443.1x, 443.2x, 443.3x, 443.4x, 443.5x, 443.6x, 443.7x, 443.8x, 443.9x, 447.1x, 785.4x), Cerebrovascular Disease (362.34, 430.xx, 431.xx, 432.xx, 433.xx, 434.xx, 435.xx, 436.xx, 437.0x, 437.1x, 437.9x, 438.xx, 781.4x, 784.3x, 997.0x), Dementia (290.xx, 331.0x, 331.1x, 331.2x), Chronic Pulmonary Disease (415.0x, 416.8x, 416.9x, 491.xx, 492.xx, 493.xx, 494.xx, 496.xx), Rheumatologic Disease (710.xx, 714.xx), Peptic Ulcer Disease (531.xx, 532.xx, 533.xx, 534.xx), Mild Liver Disease (571.2x, 571.5x, 571.6x, 571.8x, 571.9x), Mild to Moderate Diabetes (250.0x, 250.1x, 250.2x, 250.3x), Chronic Diabetes (250.4x, 250.5x, 250.6x, 250.7x, 250.8x, 250.9x), Hemiplegia (342.xx, 344.xx), Renal Disease (585.xx, 586.xx, V42.0x, V45.1x, V56.xx), Malignancy (including lymphoma and leukemia) (14x.xx, 15x.xx, 16x.xx, 170.xx, 171.xx, 174.xx, 175.xx, 176.xx, 177.xx, 178.xx, 179.xx, 18x.xx, 190.xx, 191.xx, 192.xx, 193.xx, 194.xx, 195.xx, 200.xx, 201.xx, 202.xx, 203.xx, 204.xx, 205.xx, 206.xx, 207.xx, 208.xx, 273.0x, 273.3x, V10.46), Liver Disease (572.2z, 572.3z, 572.4z, 456.0z, 456.1z, 456.2z), Metastatic Solid Tumor (196.xx, 197.xx, 198.xx, 199.zz), and AIDS (042.xx, 043.xx, 044.zz). Certain diagnoses were weighted to be more 'unhealthy' than others: HIV (*6), Metastatic Solid Tumor (*6), Liver Disease (*3), Malignancy (*2), Renal Disease (*2), Hemiplegia (*2), and Chronic Diabetes (*2).

- There was a 67% increased risk of mortality in the F/BL group compared to the M/BL group (ICU: OR=1.67, 95% CI: 1.26–2.21, p-value=0.0004)
- Patients administered F/BL in the ICU setting had a 38% increased risk of a CABP related rehospitalisation compared to the M/BL patients (OR=1.38, 95% CI: 1.15–1.65, p-value=0.0004)
- A comparison of rates of any treatment related adverse event in the ICU setting showed an 8% increased risk among the F/BL patients relative to M/BL patients (OR=1.08, 95% CI: 1.00–1.16, p-value=0.0616)
 - F/BL patients had a 65% increased risk of *clostridium difficile* compared to M/BL patients (OR=1.65, 95% CI: 1.06–2.56, p-value=0.0254)
 - F/BL patients had a 12% increased risk of experiencing hematologic toxicities compared to M/BL patients (OR=1.12, 95% CI: 1.03–1.22, p-value=0.0069)
- Patients administered F/BL had, on average, stayed in hospital for almost a day longer than the M/BL patients (Mean=0.84 days, 95% CI: 0.70–0.97, p-value<0.0001)

Figure 2a: Risk of in-hospital mortality, CABP related rehospitalisations at 30 days post index CABP episode, in-hospital rate of any adverse event – F/BL vs. M/BL*

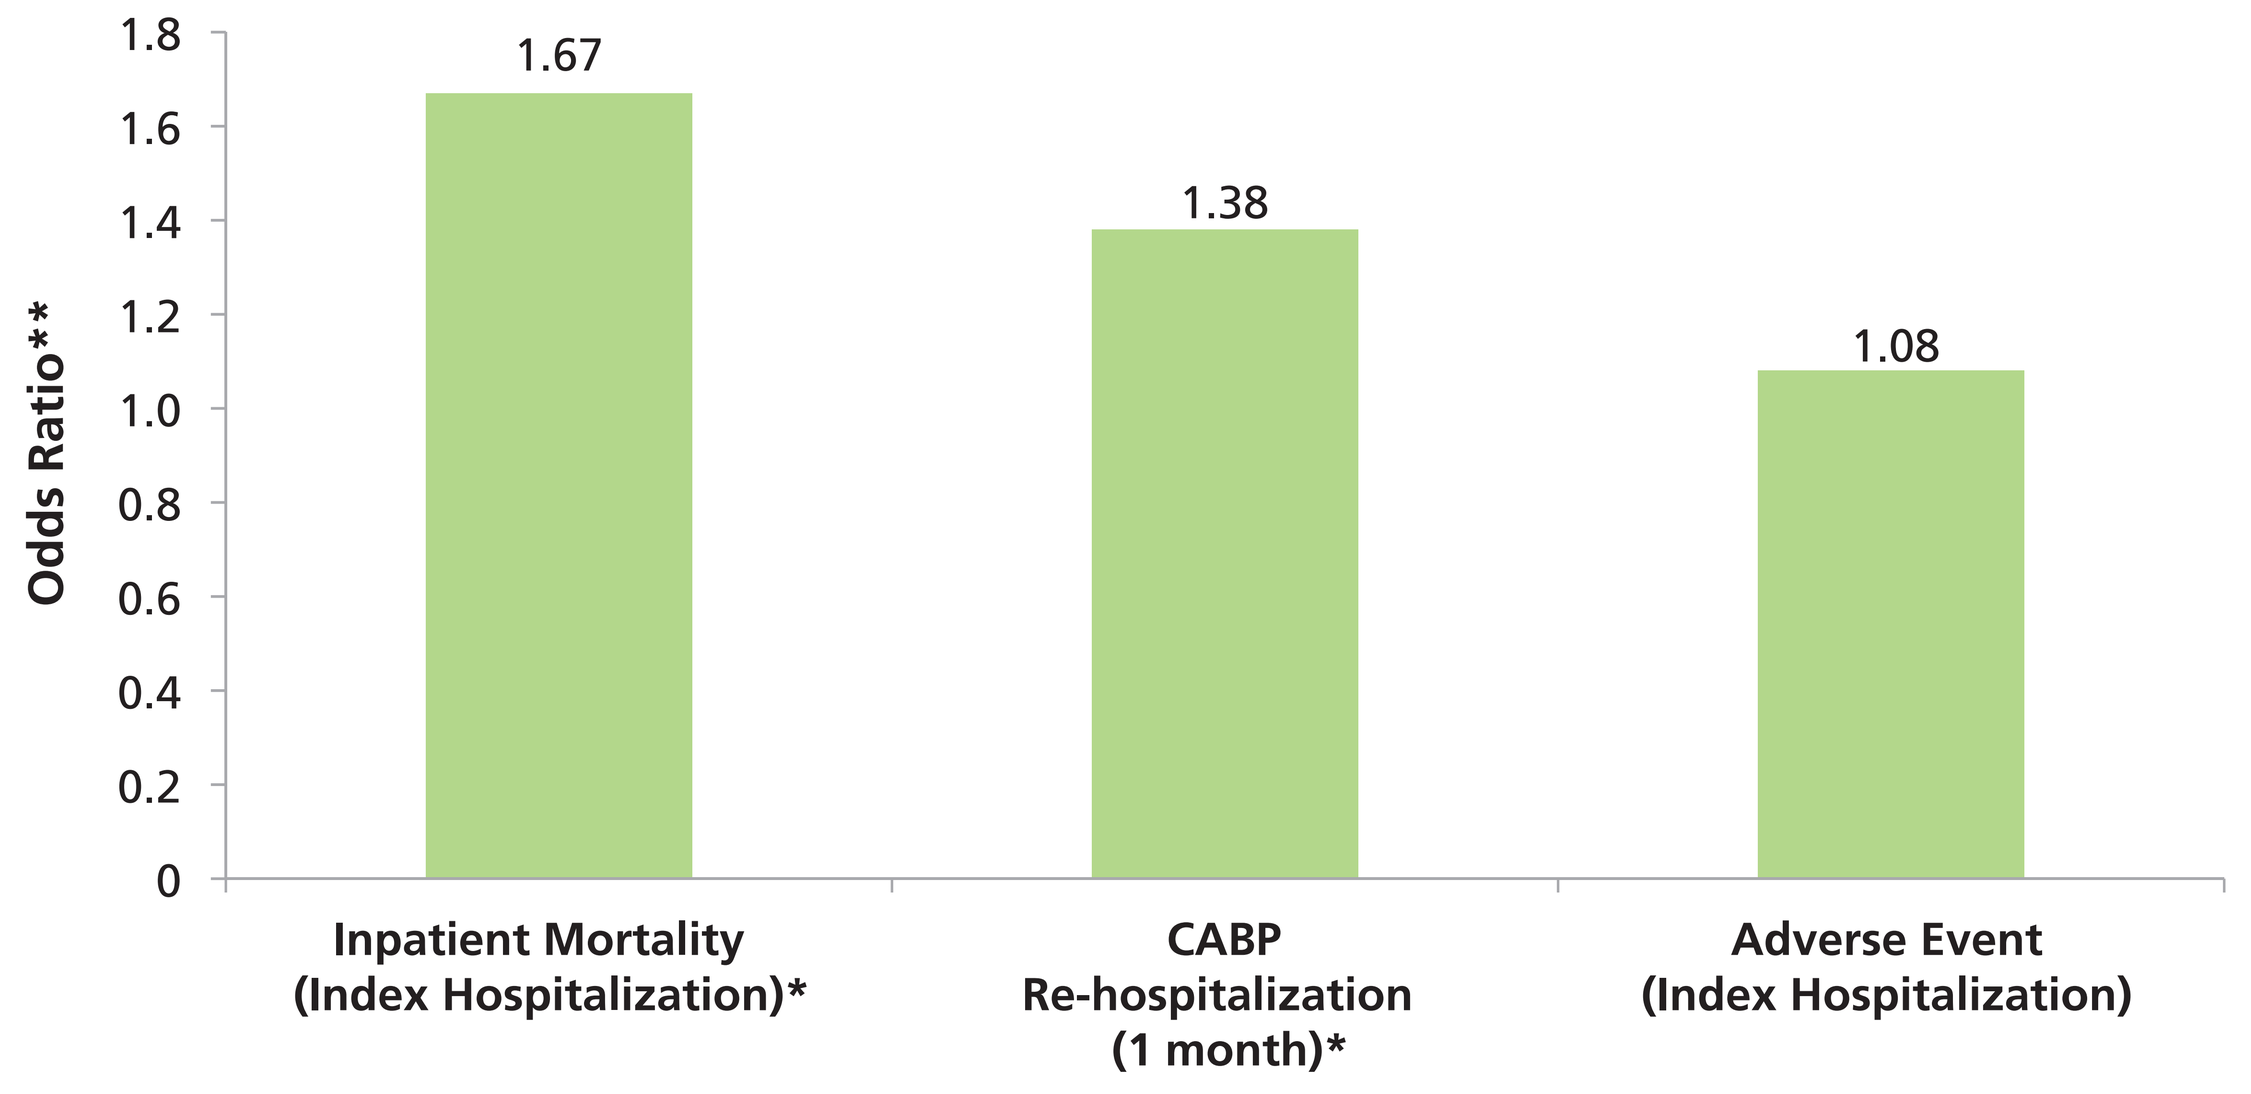
Odds Ratio**
1.8
1.6
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0
1.67
1.38
1.08
Inpatient Mortality (Index Hospitalization)*
CABP Re-hospitalization (1 month)*
Adverse Event (Index Hospitalization)

* P-value<0.05 for inpatient mortality and rehospitalisation. P-value<0.01 for adverse event.

** Adjusted Odds Ratio calculated using logistic regression, adjusting for Age, Gender, Health Index, and Length of Stay. A ratio above 1 indicates a greater risk attributed to the F/BL treatment.

Figure 2b: Risk of in-hospital rate of adverse events – F/BL vs. M/BL*

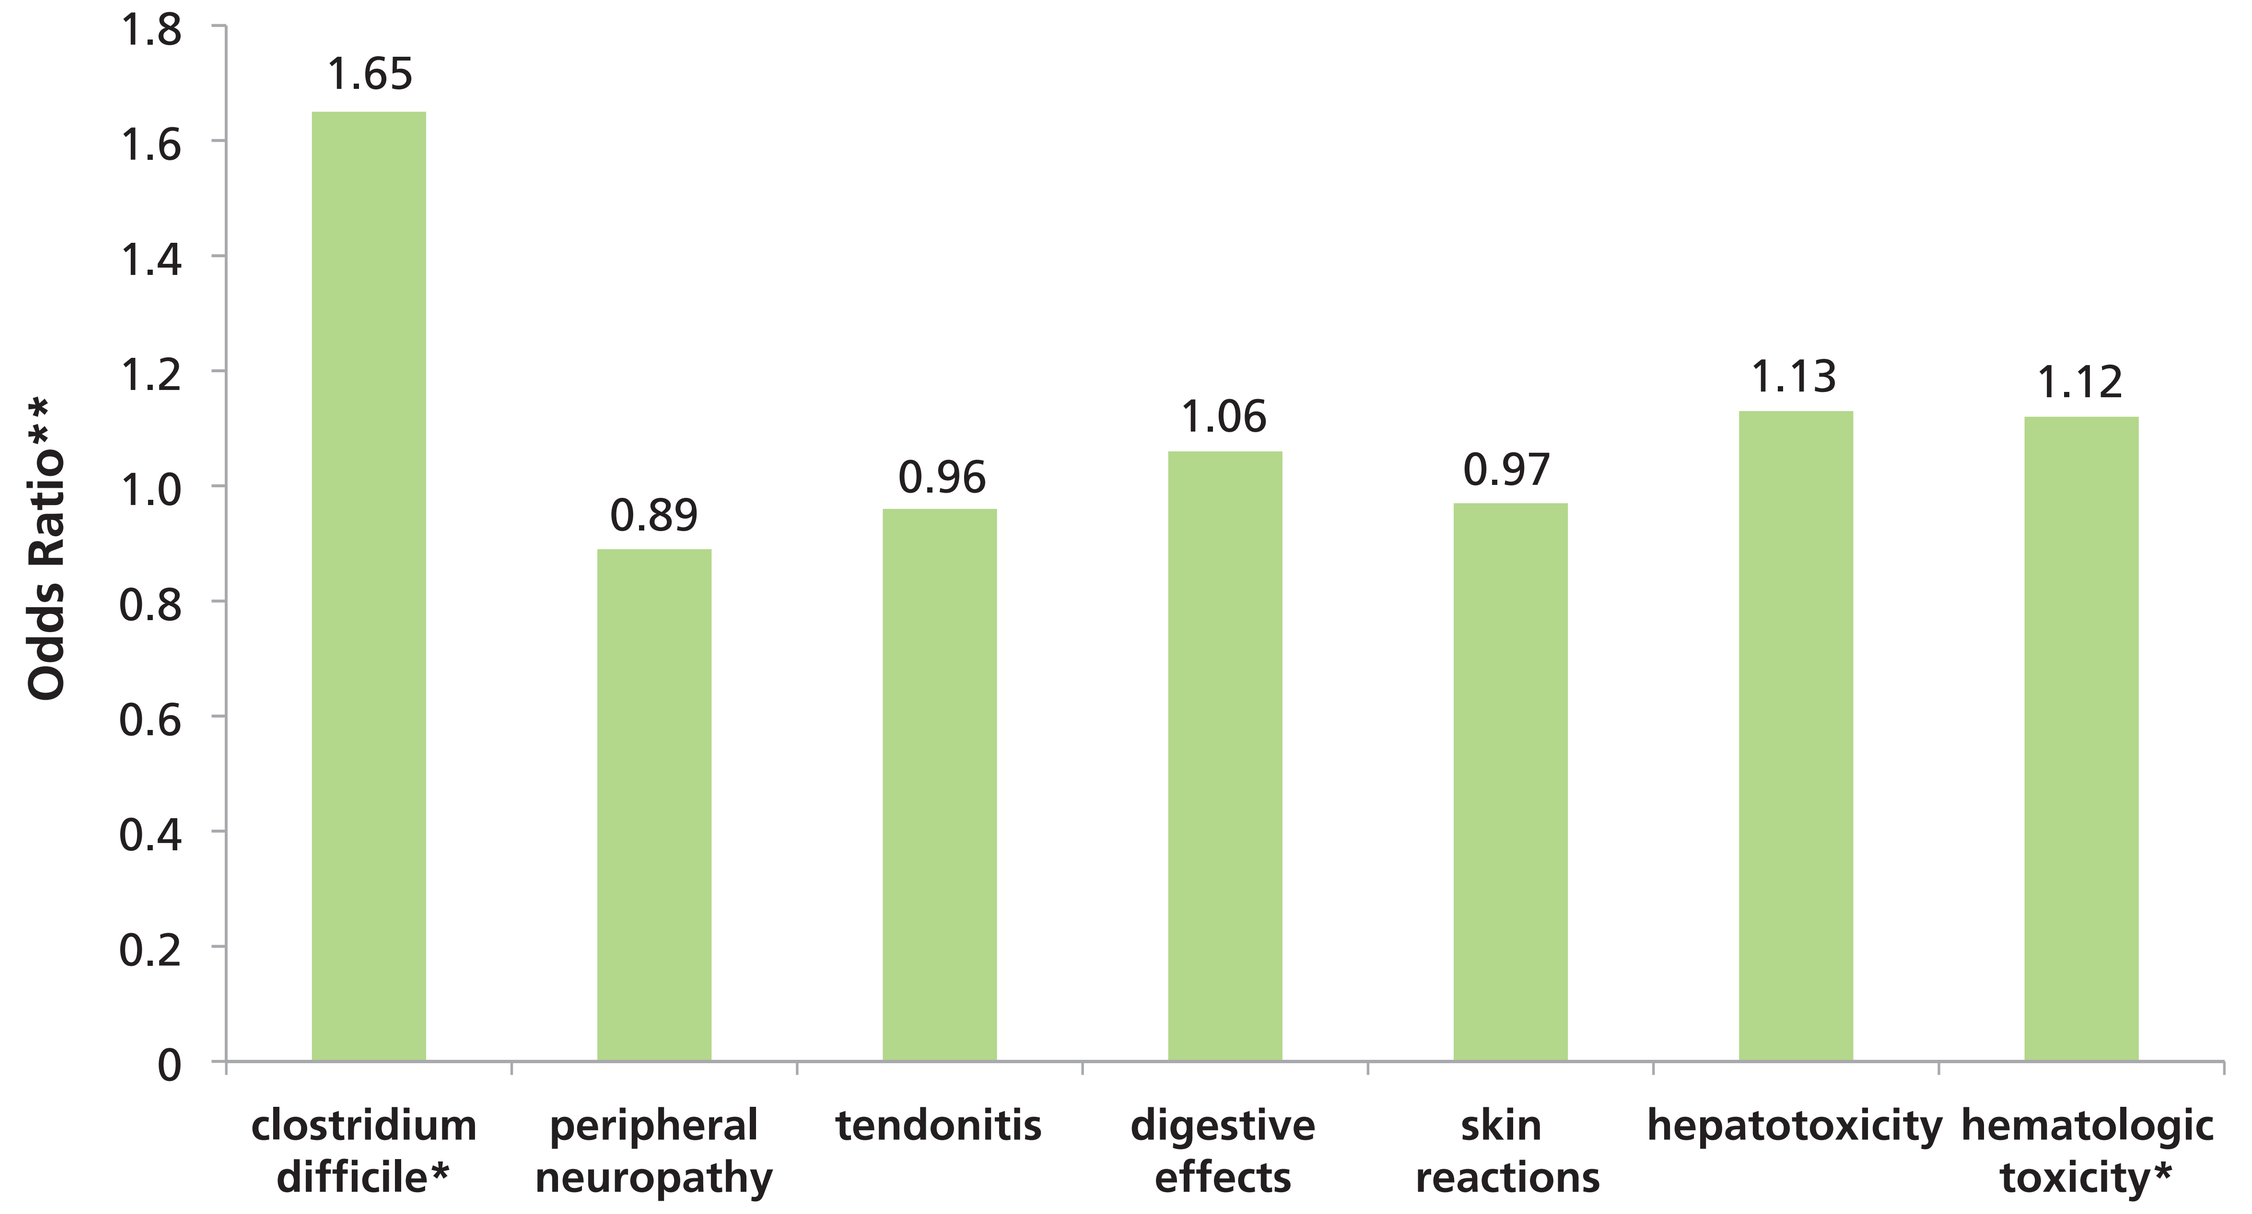
Odds Ratio**
1.8
1.6
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0
1.65
0.89
0.96
1.06
0.97
1.13
1.12
clostridium difficile*
peripheral neuropathy
tendonitis
digestive effects
skin reactions
hepatotoxicity
hematologic toxicity*

* P-value<0.05 for *clostridium difficile* and and hematologic toxicity. Other AEs were directionally mixed between the two treatments and not statistically significant (p>0.05).

** Adjusted Odds Ratio calculated using logistic regression, adjusting for Age, Gender, Health Index, and Length of Stay. A ratio above 1 indicates a greater risk attributed to the F/BL treatment.

Figure 3: Additional days spent in hospital during CABP episode – F/BL vs. M/BL*

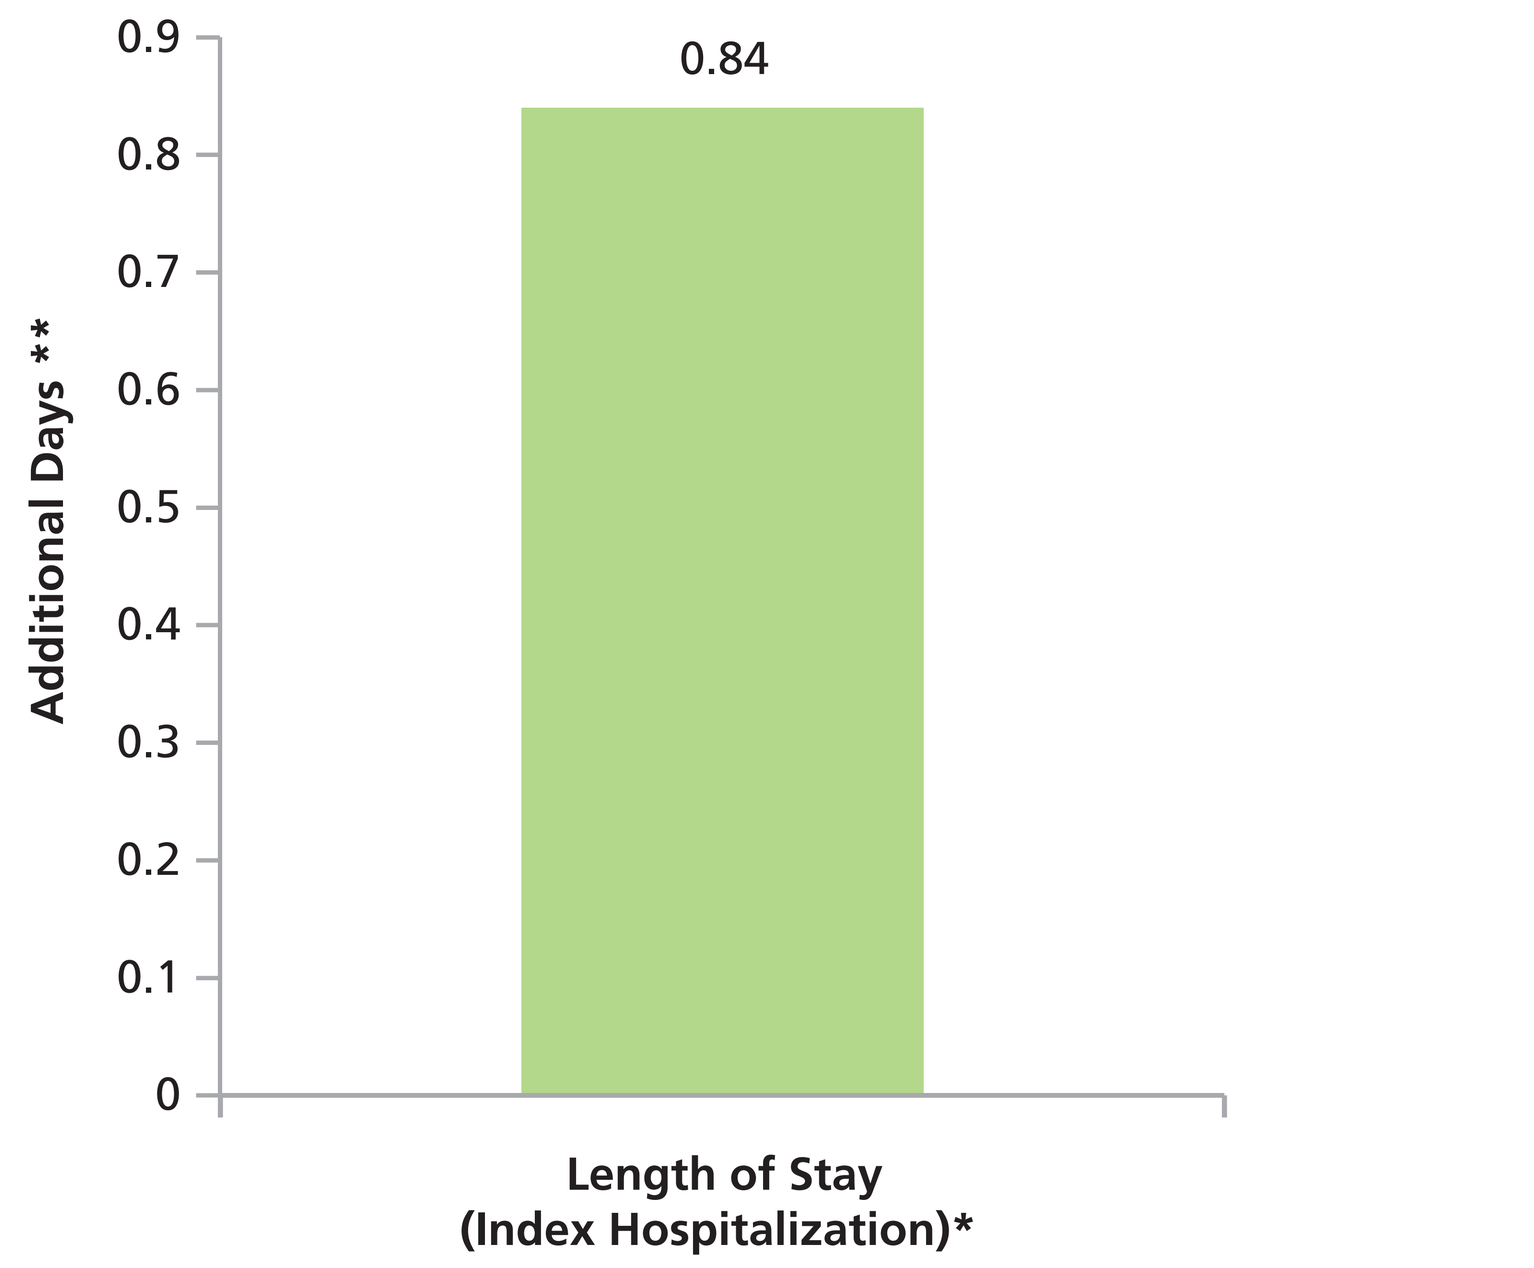
Additional Days **
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0
0.84
Length of Stay (Index Hospitalization)*

* P-value<0.05.

** Adjusted value calculated using regression adjusting for Age, Gender and Health Index.

CONCLUSIONS

- This study provides detailed, comprehensive data for examining hospital care, including drug utilization among hospitalised CABP patients
- 41% of the ICU sample were treated according to guidelines[7]
 - In the guidelines-indicated cohort, commonly used CABP treatment regimens are levofloxacin + ceftriaxone sodium and azithromycin + ceftriaxone sodium
 - The cohort receiving other treatments contained a mix including FQ monotherapy, BL monotherapy, and vancomycin in various combinations
- In the guidelines-indicated cohort, fluoroquinolone and macrolide based regimens appear to be associated with differences in outcomes that persist even when patient differences are accounted for
 - CABP patients treated with a fluoroquinolone + beta lactam showed an increased risk of treatment-related in-hospital adverse event (*clostridium difficile* and hematologic toxicities), in-hospital mortality and CABP related re-hospitalisation within 30 days of index admission, when compared to patients treated with a macrolide + beta lactam
 - CABP patients treated with a fluoroquinolone + beta lactam also on average had longer lengths of stay compared to patients treated with a macrolide + beta lactam
- A limitation of the study is the difficulty in ruling out other potential drivers of the results. Particularly, since the database is derived from billings systems, it is difficult to properly account for disease severity.

Presented at the 24th European Congress of Clinical Microbiology and Infectious Diseases, May 10–13, 2014

Research support was provided to Analysis Group, Inc. by Cempra, Inc.