P-1583

Prevalence of SCC*mec* Types and Solithromycin Susceptibility of Methicillin-resistant *Staphylococcus aureus* (MRSA) from Respiratory Samples Collected in 2012-13

Ian Morrissey*[1], Prabhavathi Fernandes[2], Barbara Lemos[1], Stephen P. Hawser[1]
[1]IHMA Europe, Sárl, Epalinges, Switzerland.
[2] Cempra, Inc., Chapel Hill, USA
*Corresponding Author

IHMA Europe, Sárl
9A Route de la Corniche,
Epalinges 1066,
Switzerland
Email:
imorrissey@ihmainc.com
Tel: +44 (0)1279 724929

Revised Abstract

Objectives: Solithromycin is a fourth generation macrolide, the first fluoroketolide being developed in oral and intravenous formulations, that is currently undergoing Phase III clinical development for the treatment of community-acquired bacterial pneumonia. This study evaluated the prevalence of SCC*mec* types associated with MRSA causing respiratory infections in 2012-13 and the *in vitro* activity of solithromycin (SOL) and comparators against these isolates.

Methods: A total of 412 respiratory MRSA were evaluated from Europe (175), North America (169), Asia-Pacific (24) and the rest of the world (44). MRSA were re-identified in a central laboratory and SCC*mec* type determined by PCR [Milheirico *et al Antimicrob Agents Chemother* 2007 51:3374]. MIC for solithromycin and comparators was determined by CLSI broth microdilution methodology.

Results: The most common SCC*mec* was IV (199 MRSA, 48.3%), followed by II (112, 27.2%), III (52, 12.6%), I (37, 9.0%) and V (3, 0.7%). Nine MRSA (2.2%) were novel SCC*mec* types. MIC_{50} for azithromycin (AZI), levofloxacin (LFX), SOL, trimethoprim-sulfamethoxazole (SXT) and tigecycline (TGC) against the major SCC*mec* types are shown in the Table. AZI and LFX were inactive against all MRSA irrespective of SCC*mec*. SXT and TGC were equally active against all MRSA. SOL was active against the most common SCC*mec* type IV and type III but not I or II.

SCC*mec* type	MIC_{50} (mg/L):				
	SOL	AZI	LFX	SXT	TGC
All (412)	16	>4	>2	<=0.5	0.25
IV (199)	0.12	>4	>2	<=0.5	0.12
II (112)	>32	>4	>2	<=0.5	0.25
III (52)	0.12	>4	>2	<=0.5	0.25
I (37)	>32	>4	>2	<=0.5	0.25

Conclusions: As SCC*mec* IV is linked with community-associated isolates of MRSA, whereas types I and II are associated with hospital-acquired MRSA, these data suggest that solithromycin may be suitable for the treatment of community-associated respiratory infections caused by MRSA.

Introduction

Solithromycin is a fourth generation macrolide, the first fluoroketolide being developed in oral and intravenous formulations, that is currently undergoing Phase III clinical development for the treatment of community-acquired bacterial pneumonia [1]. The staphylococcal cassette chromosome is a defining feature of MRSA, which are divided into several types. SCC*mec* Type I-III are characteristic of hospital-associated (HA)-MRSA while type IV is linked to community-associated-MRSA [2].

This study evaluated the prevalence of SCC*mec* types associated with 412 MRSA causing respiratory infections collected from various part of the world in 2012-13. *In vitro* activity of solithromycin and comparators against these isolates was determined and correlated with *SCCmec* types.

Materials & Methods

- A total of 412 respiratory MRSA were evaluated from Europe, North America, Asia-Pacific and the rest of the world (Table 1). Isolates were identified to the species level and minimum inhibitory concentrations (MICs) determined at a central testing laboratory (IHMA Europe, located in Epalinges, Switzerland).
- MICs were determined by the Clinical and Laboratory Standards Institute (CLSI) recommended broth microdilution testing method using panels prepared at IHMA [3].
- Quality controls were performed on each day of testing using appropriate ATCC control strains, following CLSI guidelines. Results were included in the analysis only when corresponding QC results were within the acceptable ranges [4].
- SCC*mec* type was determined by PCR as previously described [5]
- The 'D-test' to assess for inducible clindamycin-resistance was performed following the CLSI recommended method by broth microdilution (4 mg/L erythromycin & 0.5 mg/L clindamycin) [4]

Results

- The prevalence of SCC*mec* types associated with the 412 MRSA tested in this study is shown in Figures 1 and 2.
- Figure 3 shows the relationship between SCC*mec* type and macrolide-inducible resistance to clindamycin.
- Summary of MIC data for solithromycin and comparators against MRSA isolated from Europe, North-America, Asia-Pacific and the rest of the world is given in Table 2.
- The cumulative percentage MIC distributions for solithromycin are compared by world region and by SCC*mec* type in Figures 4 and 5.

Table 1: Number of isolates investigated and region of origin.

Region	No. MRSA isolates
Europe	175
North-America	169
Asia	24
Rest of the world*	44
TOTAL	412

*Rest of the world = Africa, Latin America, Middle East

Figure 1. SCC*mec* types characterized from 412 MRSA isolates causing respiratory infection.

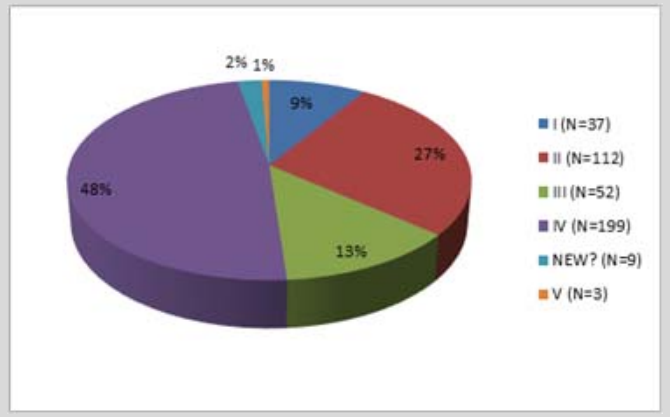
2%
1%
9%
27%
13%
48%
I (N=37)
II (N=112)
III (N=52)
IV (N=199)
NEW? (N=9)
V (N=3)

Figure 2. Percentage of SCC*mec* types characterized from MRSA causing respiratory infection in different regions of the world.

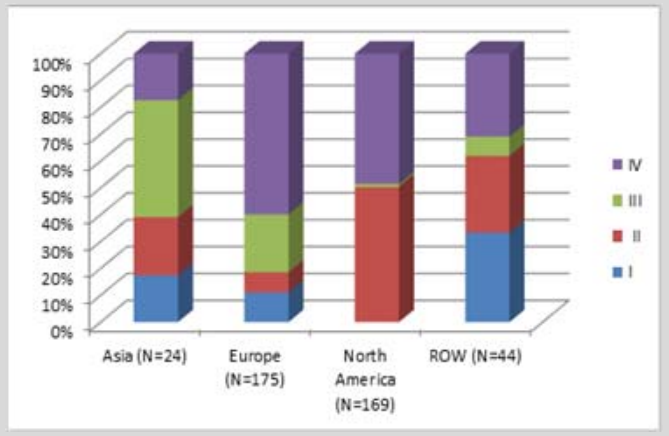
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Asia (N=24)
Europe (N=175)
North America (N=169)
ROW (N=44)
IV
III
II
I

ROW, rest of the world.

Figure 3. Percentage of macrolide-inducible resistance to clindamycin for different SCC*mec* types.

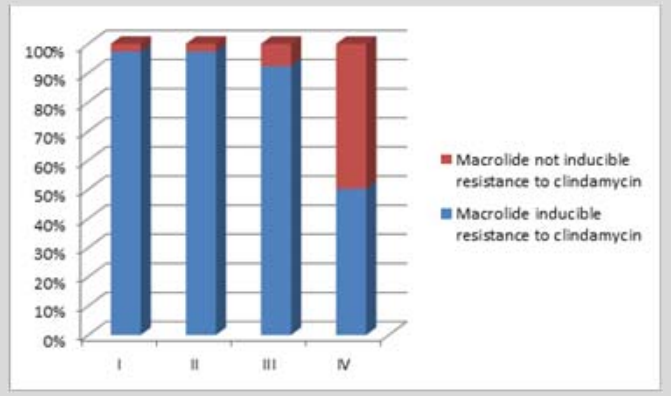
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
I
II
III
IV
Macrolide not inducible resistance to clindamycin
Macrolide inducible resistance to clindamycin

Table 2: Summary MIC data for solithromycin and comparators against 412 MRSA by SCC*mec* type.

Antimicrobial agent	SCC*mec* type (N)	MIC (mg/L)			
		50%	90%	MIN	MAX
Solithromycin	All (412)	16	>32	0.03	>32
	I (37)	>32	>32	0.06	>32
	II (112)	>32	>32	0.06	>32
	III (52)	0.12	>32	0.06	>32
	IV (199)	0.12	>32	0.03	>32
Azithromycin	All (412)	>4	>4	0.5	>4
	I (37)	>4	>4	1	>4
	II (112)	>4	>4	1	>4
	III (52)	>4	>4	0.5	>4
	IV (199)	>4	>4	0.5	>4
Daptomycin	All (412)	0.5	1	0.25	1
	I (37)	0.5	1	0.5	1
	II (112)	0.5	0.5	0.25	1
	III (52)	0.5	1	0.5	1
	IV (199)	0.5	1	0.25	1
Erythromycin	All (412)	>4	>4	0.25	>4
	I (37)	>4	>4	0.25	>4
	II (112)	>4	>4	1	>4
	III (52)	>4	>4	0.25	>4
	IV (199)	>4	>4	0.25	>4
Levofloxacin	All (412)	>2	>2	0.12	>2
	I (37)	>2	>2	0.25	>2
	II (112)	>2	>2	0.25	>2
	III (52)	>2	>2	0.25	>2
	IV (199)	>2	>2	0.12	>2
Linezolid	All (412)	2	2	≤0.5	>8
	I (37)	2	2	1	2
	II (112)	2	2	1	>8
	III (52)	2	2	1	4
	IV (199)	2	2	≤0.5	4
Telithromycin	All (412)	>2	>2	≤0.03	>2
	I (37)	>2	>2	0.12	>2
	II (112)	>2	>2	0.06	>2
	III (52)	0.25	>2	0.06	>2
	IV (199)	0.12	>2	≤0.03	>2
Tigecycline	All (412)	0.12	0.5	0.03	>2
	I (37)	0.25	0.5	0.12	1
	II (112)	0.25	0.5	0.03	>2
	III (52)	0.25	0.5	0.12	1
	IV (199)	0.12	0.5	0.06	0.5
Trimethoprim sulfamethoxazole	All (412)	≤0.5	1	≤0.5	>32
	I (37)	≤0.5	1	≤0.5	4
	II (112)	≤0.5	1	≤0.5	32
	III (52)	≤0.5	>32	≤0.5	>32
	IV (199)	≤0.5	≤0.5	≤0.5	>32
Vancomycin	All (412)	1	1	0.5	2
	I (37)	1	1	0.5	1
	II (112)	1	1	0.5	2
	III (52)	1	1	0.5	2
	IV (199)	1	1	0.5	2

Figure 4: Cumulative percentage MIC distribution for solithromycin against MRSA causing respiratory infection in different regions of the world.

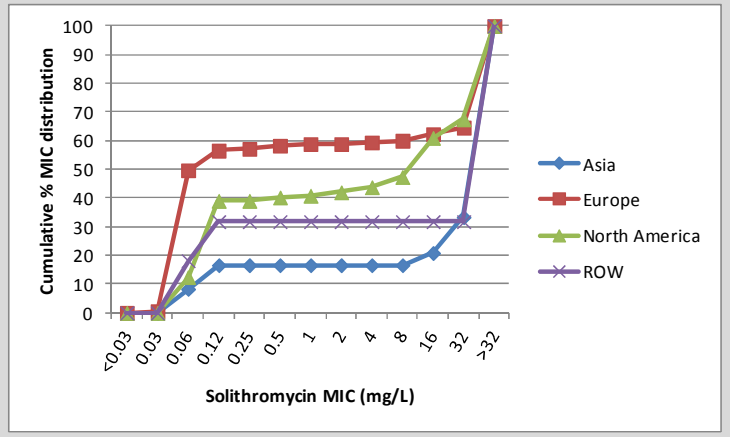
Cumulative % MIC distribution
100
90
80
70
60
50
40
30
20
10
0
<0.03
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
>32
Solithromycin MIC (mg/L)
Asia
Europe
North America
ROW

Figure 5: Cumulative percentage MIC distribution for solithromycin against MRSA causing respiratory infection with different SCC*mec* types.

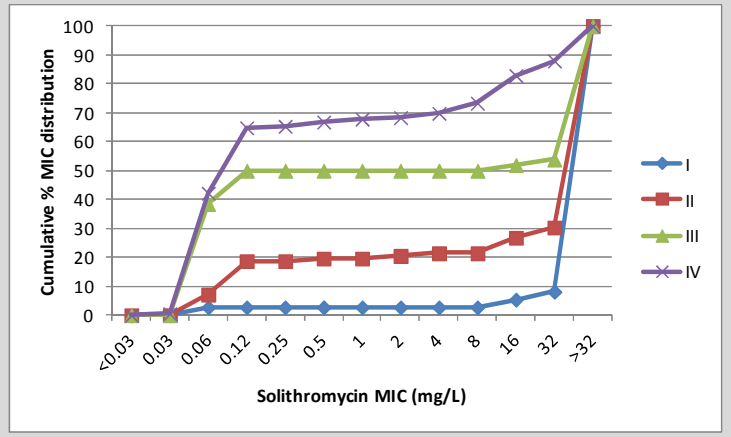
Cumulative % MIC distribution
100
90
80
70
60
50
40
30
20
10
0
<0.03
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
>32
Solithromycin MIC (mg/L)
I
II
III
IV

Conclusions

- Solithromycin was active against the majority of MRSA with SCC*mec* IV (MIC 50% = 0.12 mg/L) which was the major SCC*mec* type in Europe and highly represented in isolates from North America.
- Solithromycin was also very active against SCC*mec* type III (MIC 50% = 0.12 mg/L) but not active against the majority of SCC*mec* type I and II (MIC 50% > 32 mg/L)
- Solithromycin was considerably more active against MRSA with SCC*mec* type III and IV than the macrolides.
- Daptomycin and vancomycin were equally active against all MRSA, whereas linezolid and tigecycline were less active against SCC*mec* type II and trimethoprim-sulfamethoxazole was less active against SCC*mec* type III MRSA. Azithromycin, erythromycin, and levofloxacin were inactive against most MRSA, irrespective of SSC*mec* type. Telithromycin showed a similar pattern of activity to that seen with solithromycin.
- SCC*mec* type IV was less associated with macrolide-inducible resistance to clindamycin than type I, II and III.
- These data show that solithromycin could be a good alternative to treat community-associated MRSA, which are commonly SCC*mec* IV, and support the continued development of this antibiotic.

References

1. Oldach D, Clark K, Schranz J, Das A, Craft JC, Scott D, Jamieson BD, Fernandes P. 2013. Randomized, *double-blind, multicenter phase* 2 study comparing the efficacy and safety of oral solithromycin (CEM-101) to those of oral levofloxacin in the treatment **of** patients with community-acquired bacterial pneumonia. Antimicrob Agents Chemother 57:2526-34.
2. Otter JA, French GL. 2010. Molecular epidemiology of community-associated methicillin-resistant *Staphylococcus aureus* in Europe. Lancet Infect Dis 10:227-239.
3. Clinical Laboratory Standards Institute. 2012. Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standards – Ninth Edition. CLSI document M07-A9. Wayne, PA.
4. Clinical and Laboratory Standards Institute. 2013. Performance Standards for Antimicrobial Susceptibility Testing; Twenty-third Informational Supplement. CLSI Document M100-S23. Wayne, PA.
5. Milheiriço C, Oliveira DC, de Lencastre H. 2007. Update to the multiplex PCR strategy for assignment of *mec* element types in *Staphylococcus aureus*. Antimicrob Agents Chemother 51:3374-7

Acknowledgements

This study was sponsored by a grant from Cempra Pharmaceuticals.