Characterization of the Mechanism of Nicotinic Acid Acetylcholine Receptor Inhibition that is Likely Linked to the Off-Target Activity by Telithromycin

Daniel Bertrand[1], Sonia Bertrand[1], David Pereira[2], Kara Keedy[2], *Prabhavathi Fernandes[2]

[1]HiQScreen Sàrl, 15, rue de l'Athénée, Geneva Switzerland; [2]Cempra Pharmaceuticals Inc. Chapel Hill, NC 27517 USA

Poster P1418

ABSTRACT

Objectives: Previous studies carried out with telithromycin at the nicotinic acetylcholine receptors have clearly illustrated that the pyridine moiety in the side-chain of telithromycin inhibits the α7 and α3β4 receptors. Similarly, the visual effects of voriconazole led us to the characterization of the inhibition of α3β4 nAChRs by its heterocyclic N in the pyrimidine side chain. The aim of this study was to examine the mode of action of telithromycin at the human α7 and α3β4 nAChRs.

Methods: Electrophysiological studies were conducted using expression of human nAChRs in *Xenopus* oocytes. ACh dose-response curves were obtained in the absence or presence of a fixed concentration of telithromycin to determine the mode of action of telithromycin. Competitive antagonists are characterized by the fact that blockade caused by the antagonist can be surmounted by the appropriate increase in the agonist concentration. On the contrary, non-competitive antagonists are characterized by the fact that blockade is insurmountable.

Results: Data obtained for α3β4 with 2 µM telithromycin suggests that telithromycin might have a dual action with competitive and non-competitive inhibition. The dual mode of action of telithromycin was confirmed by examining the time course of the ACh response measured at a low ACh concentration (10 µM) and at a high ACh concentration (1280 µM). Inhibition caused by telithromycin is not accompanied by a modification of the response time course at 10 µM ACh, whereas a profound modification of the decay time was observed at the high ACh-concentration. The difference in the response time course, with a faster decay time observed at ACh concentrations >160 µM, indicates that inhibition is not caused by competition only, but that telithromycin probably enters the channel pore and blocks ionic conduction by steric hindrance. Exposure of cells expressing the human α7 to telithromycin (20 µM) causes a shift in the concentration activation curve towards higher ACh concentrations indicative of a competitive inhibition of α7. Similarly to α3β4 at high ACh concentrations (>600 µM), telithromycin causes an additional inhibition probably due to open channel blockade.

Conclusions: Mechanistic characterization of the side effects of drugs helps to optimize the side-effect profiles of drugs in development. These studies can mechanistically differentiate new macrolides/ketolides from telithromycin.

OBJECTIVES

Macrolides are known generally as a safe class of antibiotics. Soon after it received marketing approval, telithromycin, or Kekek, was noted to cause serious side effects that included i) reversible visual disturbances, ii) loss of consciousness, iii) exacerbation of myasthenia gravis and iv) hepatotoxicity. Identification of the mechanism of these side effects is essential to development of new macrolides and ketolides in the future. Previous studies carried out with telithromycin at the nicotinic acetylcholine receptors have clearly illustrated that the pyridine moiety in the side-chain of telithromycin inhibits the α7 and α3β4 receptors. Similarly, the visual effects of voriconazole led us to the characterization of the inhibition of α3β4 nAChRs by its heterocyclic N in the pyrimidine side chain. The aim of this study was to examine the mode of action of telithromycin at the human α7 and α3β4 nAChRs.

RESULTS

Figure 1:
Nicotinic acetylcholine receptors are widely expressed in the nervous system, immune system and multiple organs

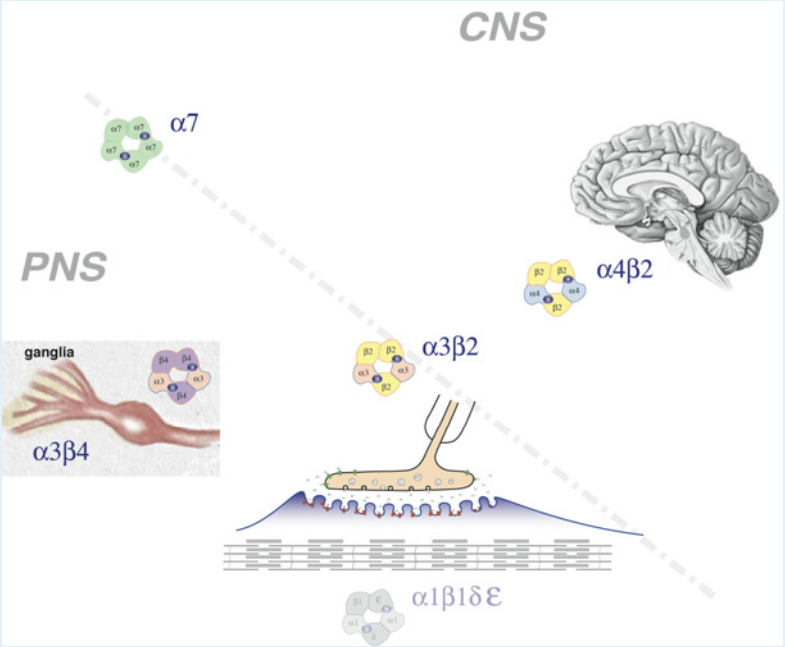
CNS
PNS
α7
α4β2
α3β2
ganglia
α3β4
α1β1δε

Schematic representation of principal nAChRs subtypes expressed in the nervous system.

Figure 2:
Concentration activation curve to ACh in absence or presence of a fixed concentration of telithromycin

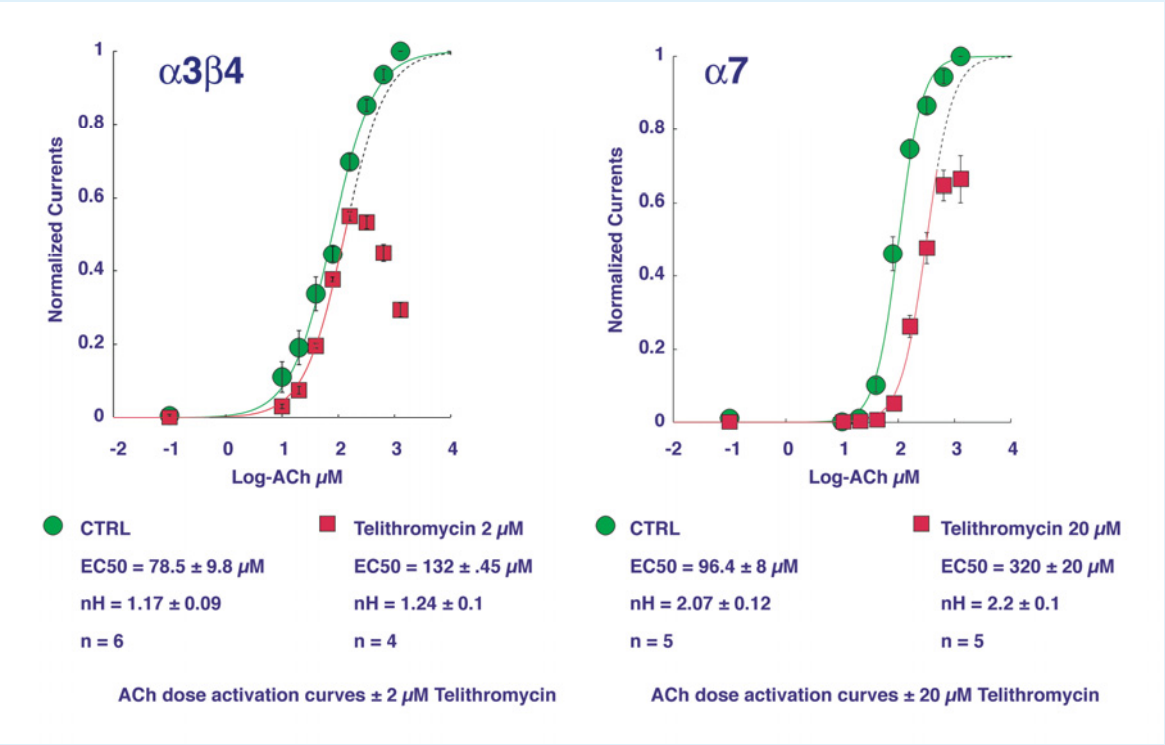
α3β4
α7
Normalized Currents
Log-ACh µM
CTRL
EC50 = 78.5 ± 9.8 µM
nH = 1.17 ± 0.09
n = 6
Telithromycin 2 µM
EC50 = 132 ± .45 µM
nH = 1.24 ± 0.1
n = 4
CTRL
EC50 = 96.4 ± 8 µM
nH = 2.07 ± 0.12
n = 5
Telithromycin 20 µM
EC50 = 320 ± 20 µM
nH = 2.2 ± 0.1
n = 5
ACh dose activation curves ± 2 µM Telithromycin
ACh dose activation curves ± 20 µM Telithromycin

At α3β4, an exposure to 2 µM telithromycin causes a shift in the $EC_{50's}$ (from 78 to 132 µM) and a break down of the curve above 160 µM ACh. Similarly, at α7 an exposure to 20 µM telithromycin causes a shift in the $EC_{50's}$ (from 96 to 320 µM) and a break down of the curve above 600 µM ACh.

This can be interpreted as a dual action of telithromycin with competitive inhibition and an open channel blockade.

Figure 3:
The dual mode of action of telithromycin at α3β4

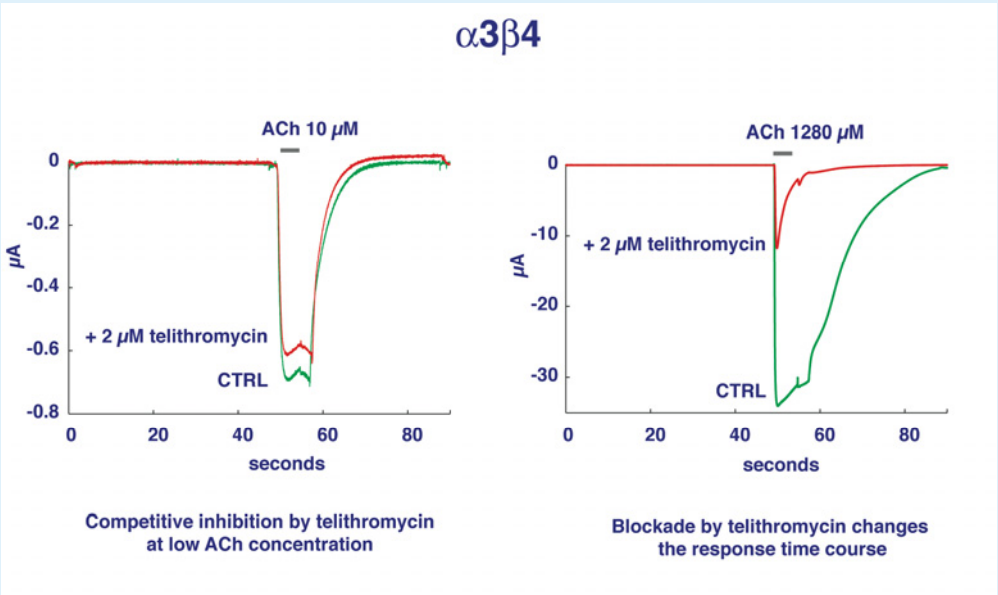
α3β4
ACh 10 µM
ACh 1280 µM
µA
+ 2 µM telithromycin
CTRL
seconds
Competitive inhibition by telithromycin at low ACh concentration
Blockade by telithromycin changes the response time course

For low ACh concentrations (< 160 µM), exposure to 2 µM telithromycin causes only a small reduction of the current amplitude without noticeable changes in the response time course. This data supports a competitive mode of action for telithromycin. The faster decay of the responses observed at high ACh concentrations is, however, indicative of a non-competitive blockade.

Figure 4:
Telithromycin changes the α7 response time course

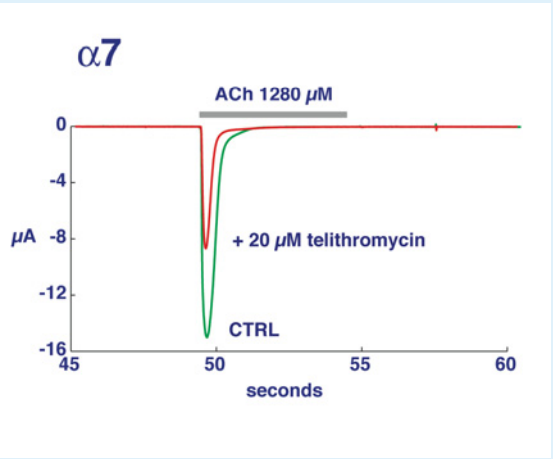
α7
ACh 1280 µM
µA
+ 20 µM telithromycin
CTRL
seconds

Data clearly illustrate the faster decay time observed in presence of telithromycin suggesting that telithromycin probably acts as an open channel blocker at high ACh concentrations.

Figure 5:
Schematic receptor representation and binding sites

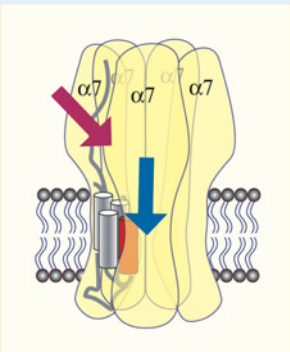
α7
α7
α7

The red arrow indicates the binding site at the interface between subunits, that forms the ligand binding domain, and blue arrow the binding site in the channel domain.

CONCLUSIONS

- **A dual mechanism of action with a competitive blockade for the low concentrations and an open channel blocker effect for the high ACh concentrations was observed both at α3β4 and α7 nAChRs.**
- **The open channel blocker effect caused by telithromycin done at high ACh concentrations revealed a modification of the decay time indicative of the non-competitive inhibition.**
- **Altogether, these data indicate that telithromycin is acting by a dual mechanism of competitive inhibition, that is binding in the orthosteric ligand binding site, and non-competitive inhibition at high ACh concentrations that is probably caused by an open channel blocker effect.**
- **Mechanistic characterization of telithromycin side effects could help to optimize drugs in development. These studies can mechanistically differentiate new macrolides/ketolides from telithromycin.**

Funding: Cempra Pharmaceuticals