Poster 467
ASM Microbe
June 18, 2016
Boston, MA

Microbiological Diagnoses from a Recent Community-acquired Bacterial Pneumonia (CABP) Trial – SOLITAIRE-IV

A. Sheets, K. Keedy, B. Jamieson, D. Oldach, P. Fernandes
Cempra, Inc., Chapel Hill, NC, USA

Amanda Sheets, PhD
6320 Quadrangle Drive, Suite 360
Chapel Hill, North Carolina 27517
Tel. 919.576.2303
asheets@cempra.com

Introduction

- Community-acquired bacterial pneumonia (CABP) is among the most common and serious infections requiring systemic antibiotic therapy[1-2] and is associated with significant morbidity and mortality, despite therapeutic advances[3].
- The emergence and spread of respiratory pathogens resistant to macrolides and other classes of antibiotics have begun to limit therapeutic options for CABP[4,5].
- A macrolide with improved activity, a better safety profile and available in both intravenous (IV) and oral formulations would be a significant therapeutic advance in the treatment of CABP.
- Solithromycin (SOLI) is a fourth-generation macrolide, with potent in vitro activity against CABP pathogens, including atypical bacteria and macrolide resistant strains, and is being developed in oral and IV formulations.

Phase 3 Trials in CABP

- Moxifloxacin (MOXI), a potent fluoroquinolone, was selected as the comparator because of its proven effectiveness in CABP and the ability to study both oral and IV formulations worldwide with a consistent dosing regimen.
- Oral solithromycin demonstrated statistical non-inferiority to oral moxifloxacin in treating adults with CABP in the outpatient setting (SOLITAIRE-Oral trial)[6].
- SOLITAIRE-IV was a global non-inferiority trial of IV-to-oral SOLI versus MOXI.

Methods

Study Design

- The SOLITAIRE-IV trial was conducted under the new FDA CABP Guidance with clinical outcomes measured using an objective endpoint of early clinical response (ECR) at 72h post-dose.
- 863 patients with confirmed CABP (PORT II to IV) were randomized between January 2014 and July 2015 to receive IV SOLI or MOXI on Day 1 and were permitted to switch to oral dosing on subsequent days.

Visit Schedule

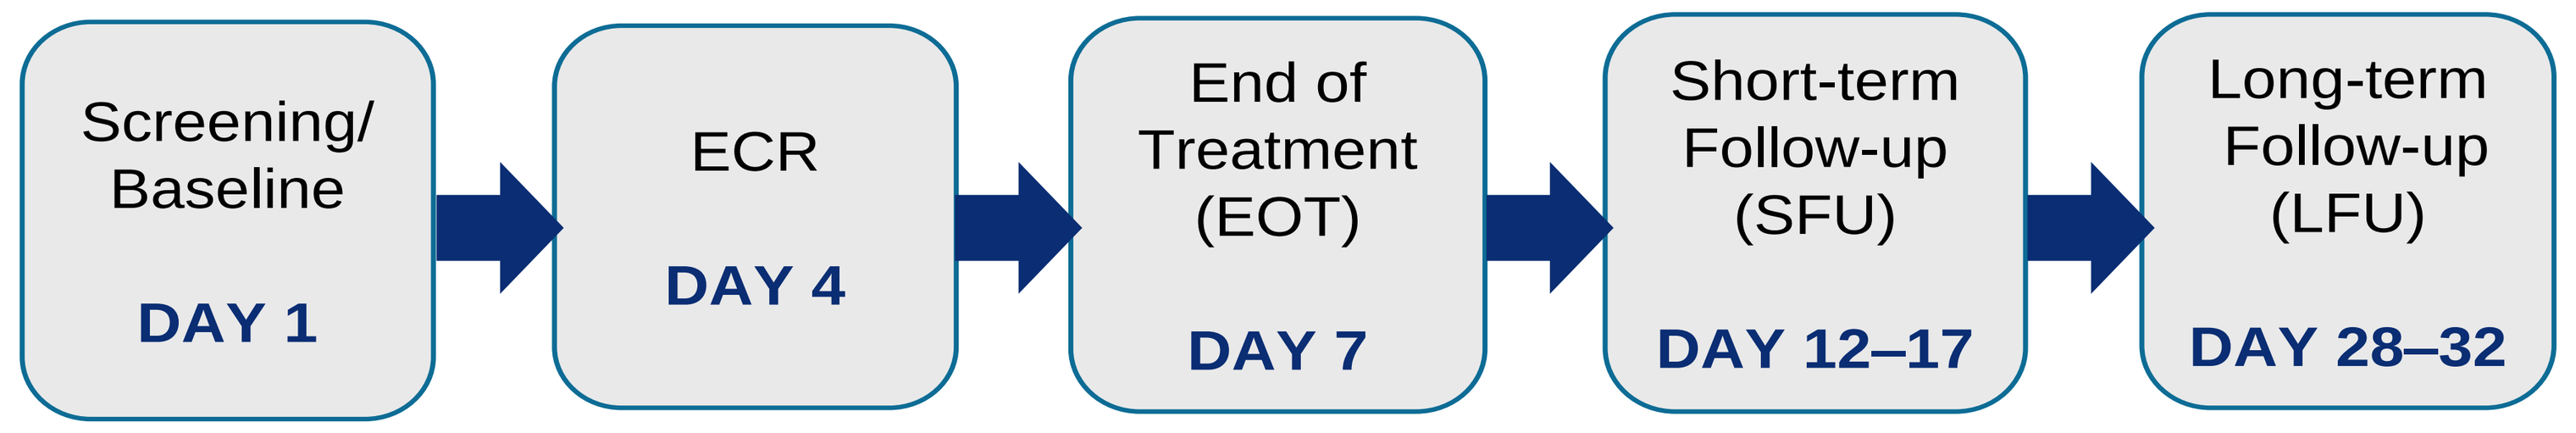
Screening/ Baseline
DAY 1
ECR
DAY 4
End of Treatment (EOT)
DAY 7
Short-term Follow-up (SFU)
DAY 12–17
Long-term Follow-up (LFU)
DAY 28–32

Microbiological Assessments

- A variety of techniques were used to enhance the detection of pathogens
 - blood and sputum culture
 - urinary antigen test (EIA) - *S. pneumoniae* and *L. pneumophila*
 - serology (4-fold rise in titer at LFU) - *L. pneumophila* and *M. pneumoniae*
 - quantitative PCR of nasopharyngeal swabs for *S. pneumoniae*
 - culture and qPCR of oropharyngeal swabs for *M. pneumoniae*

Analysis Populations

- The ITT (intent-to-treat) population consists of all randomized patients, the mITT (microbiological ITT) population consists of all randomized patients with a baseline pathogen identified, the CE (clinically evaluable) population consists of patients who met inclusion/exclusion criteria without significant protocol deviations, the ME (microbiologically evaluable) population is the intersection of the mITT and CE populations.

Efficacy Analyses

- Primary: Early clinical response (improvement of cough / dyspnea / chest pain / sputum production without worsening of any) at 72h in the ITT population
- Key Secondary: ECR in the mITT, investigators assessment of clinical response at SFU visit in the ITT/CE-SFU. Additional: clinical response at SFU in the mITT/ME-SFU. By-pathogen outcome at EOT/SFU visits in mITT/ME populations.

Results

Table 1. Early Clinical Response and Clinical Success at SFU

Outcome Measure	SOLI, % (n/N)	MOXI, % (n/N)	Delta, % (95% CI)
Early Clinical Response			
ITT Population	79.3 (344/434)	79.7 (342/429)	-0.46 (-6.1, 5.2)
mITT Population	80.3 (139/173)	79.1 (121/153)	+1.26 (-8.1, 10.6)
ME-ECR Population	83.6 (138/165)	80.3 (118/147)	+3.36 (-5.8, 12.6)
Clinical Success at SFU			
ITT Population	84.6 (367/434)	88.6 (380/429)	-4.02 (-8.8, 0.8)
mITT Population	83.2 (144/173)	88.2 (135/153)	-5.00 (-13.2, 3.2)
ME-SFU Population	86.7 (137/158)	90.1 (128/142)	-3.43 (-11.3, 4.5)

- SOLI was non-inferior to MOXI in ECR in both the ITT and mITT populations.

Results

Figure 1. Microbiological Diagnoses (mITT population)

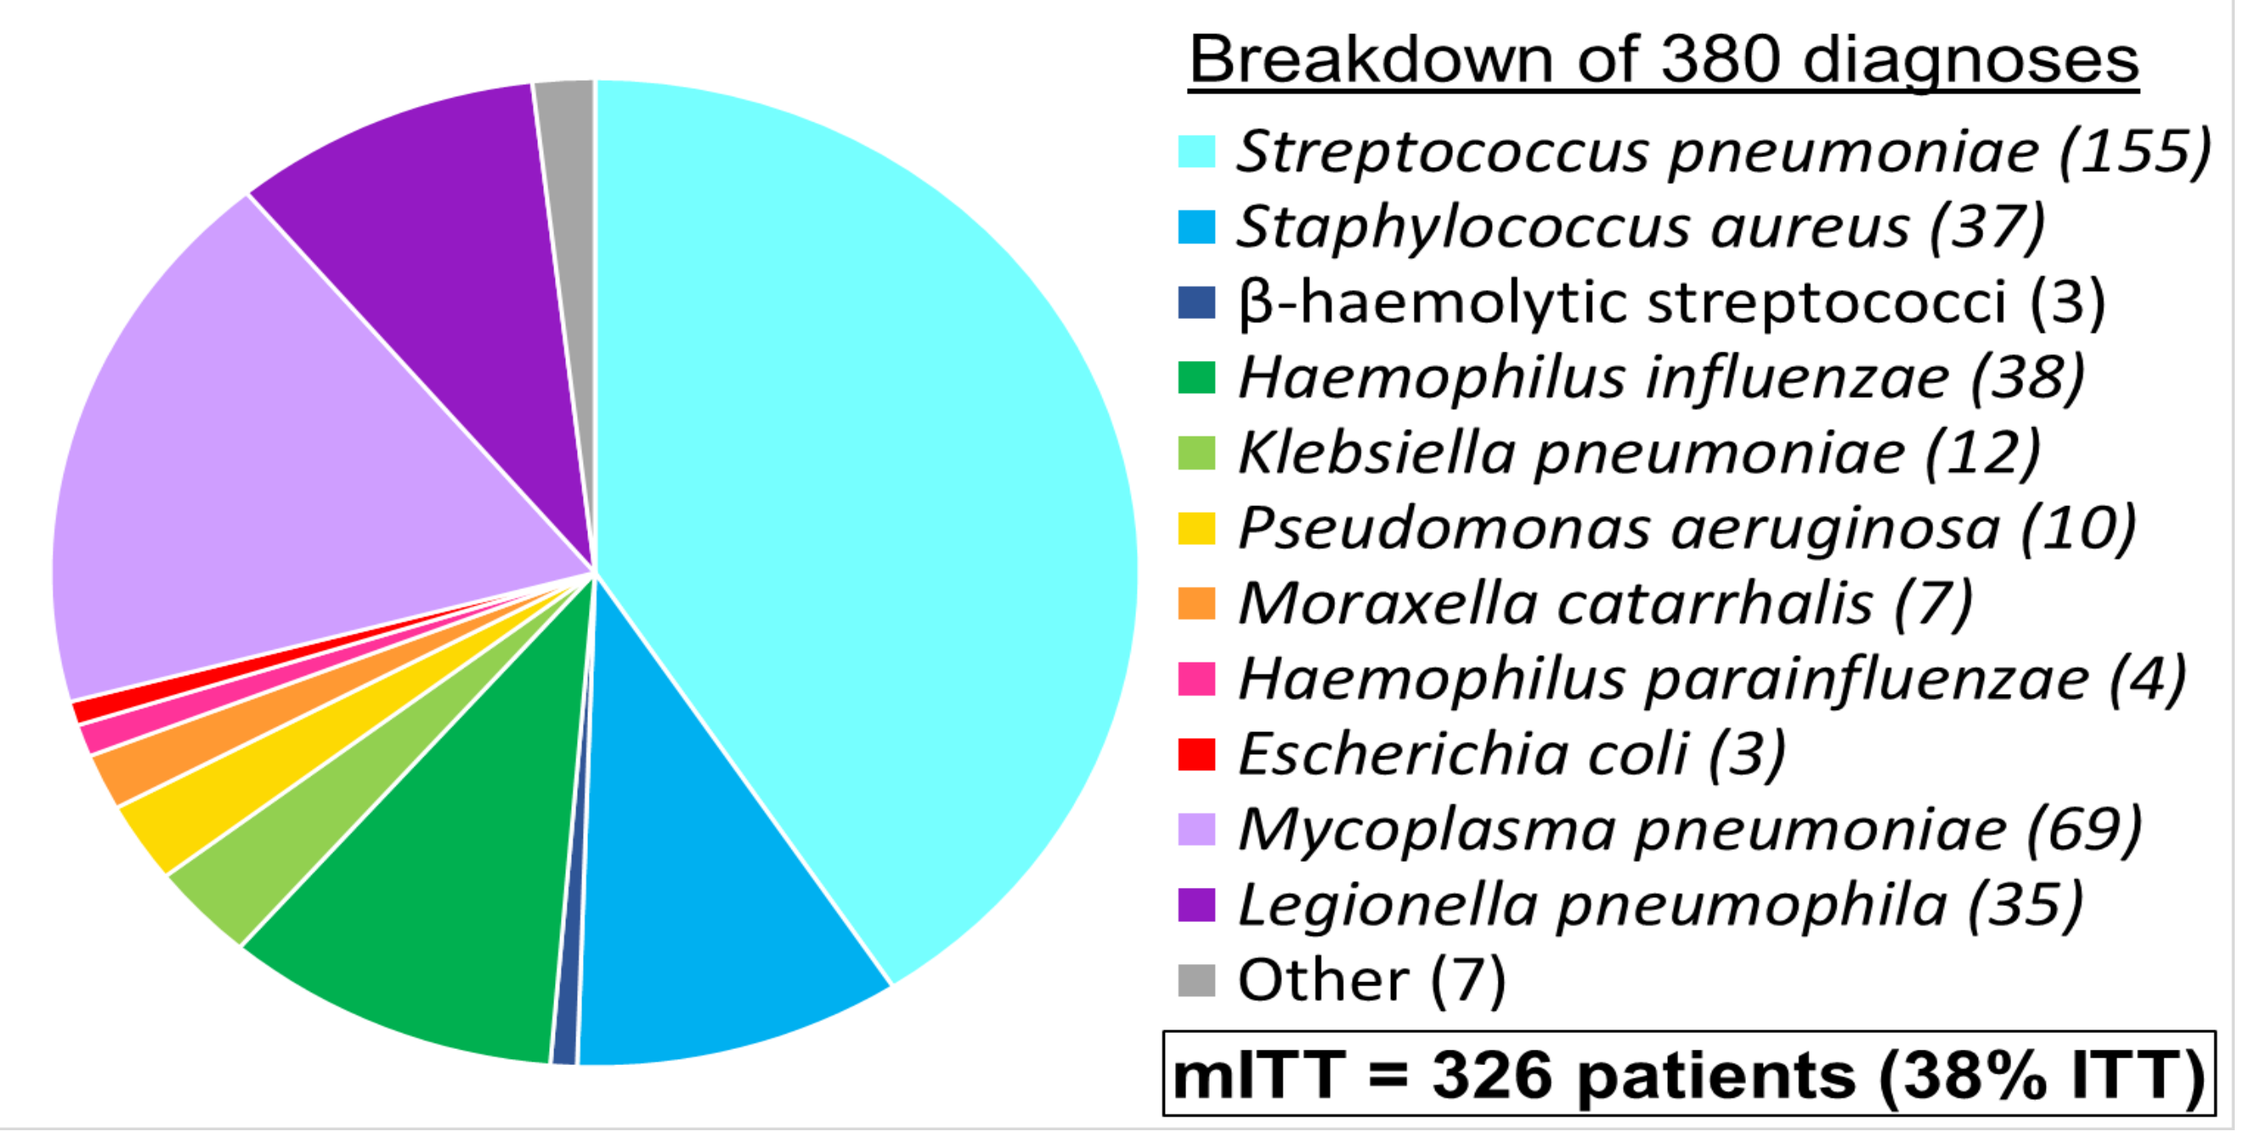
Breakdown of 380 diagnoses
Streptococcus pneumoniae (155)
Staphylococcus aureus (37)
β-haemolytic streptococci (3)
Haemophilus influenzae (38)
Klebsiella pneumoniae (12)
Pseudomonas aeruginosa (10)
Moraxella catarrhalis (7)
Haemophilus parainfluenzae (4)
Escherichia coli (3)
Mycoplasma pneumoniae (69)
Legionella pneumophila (35)
Other (7)
mITT = 326 patients (38% ITT)

Figure 2. Macrolide MIC distribution for all *S. pneumoniae*

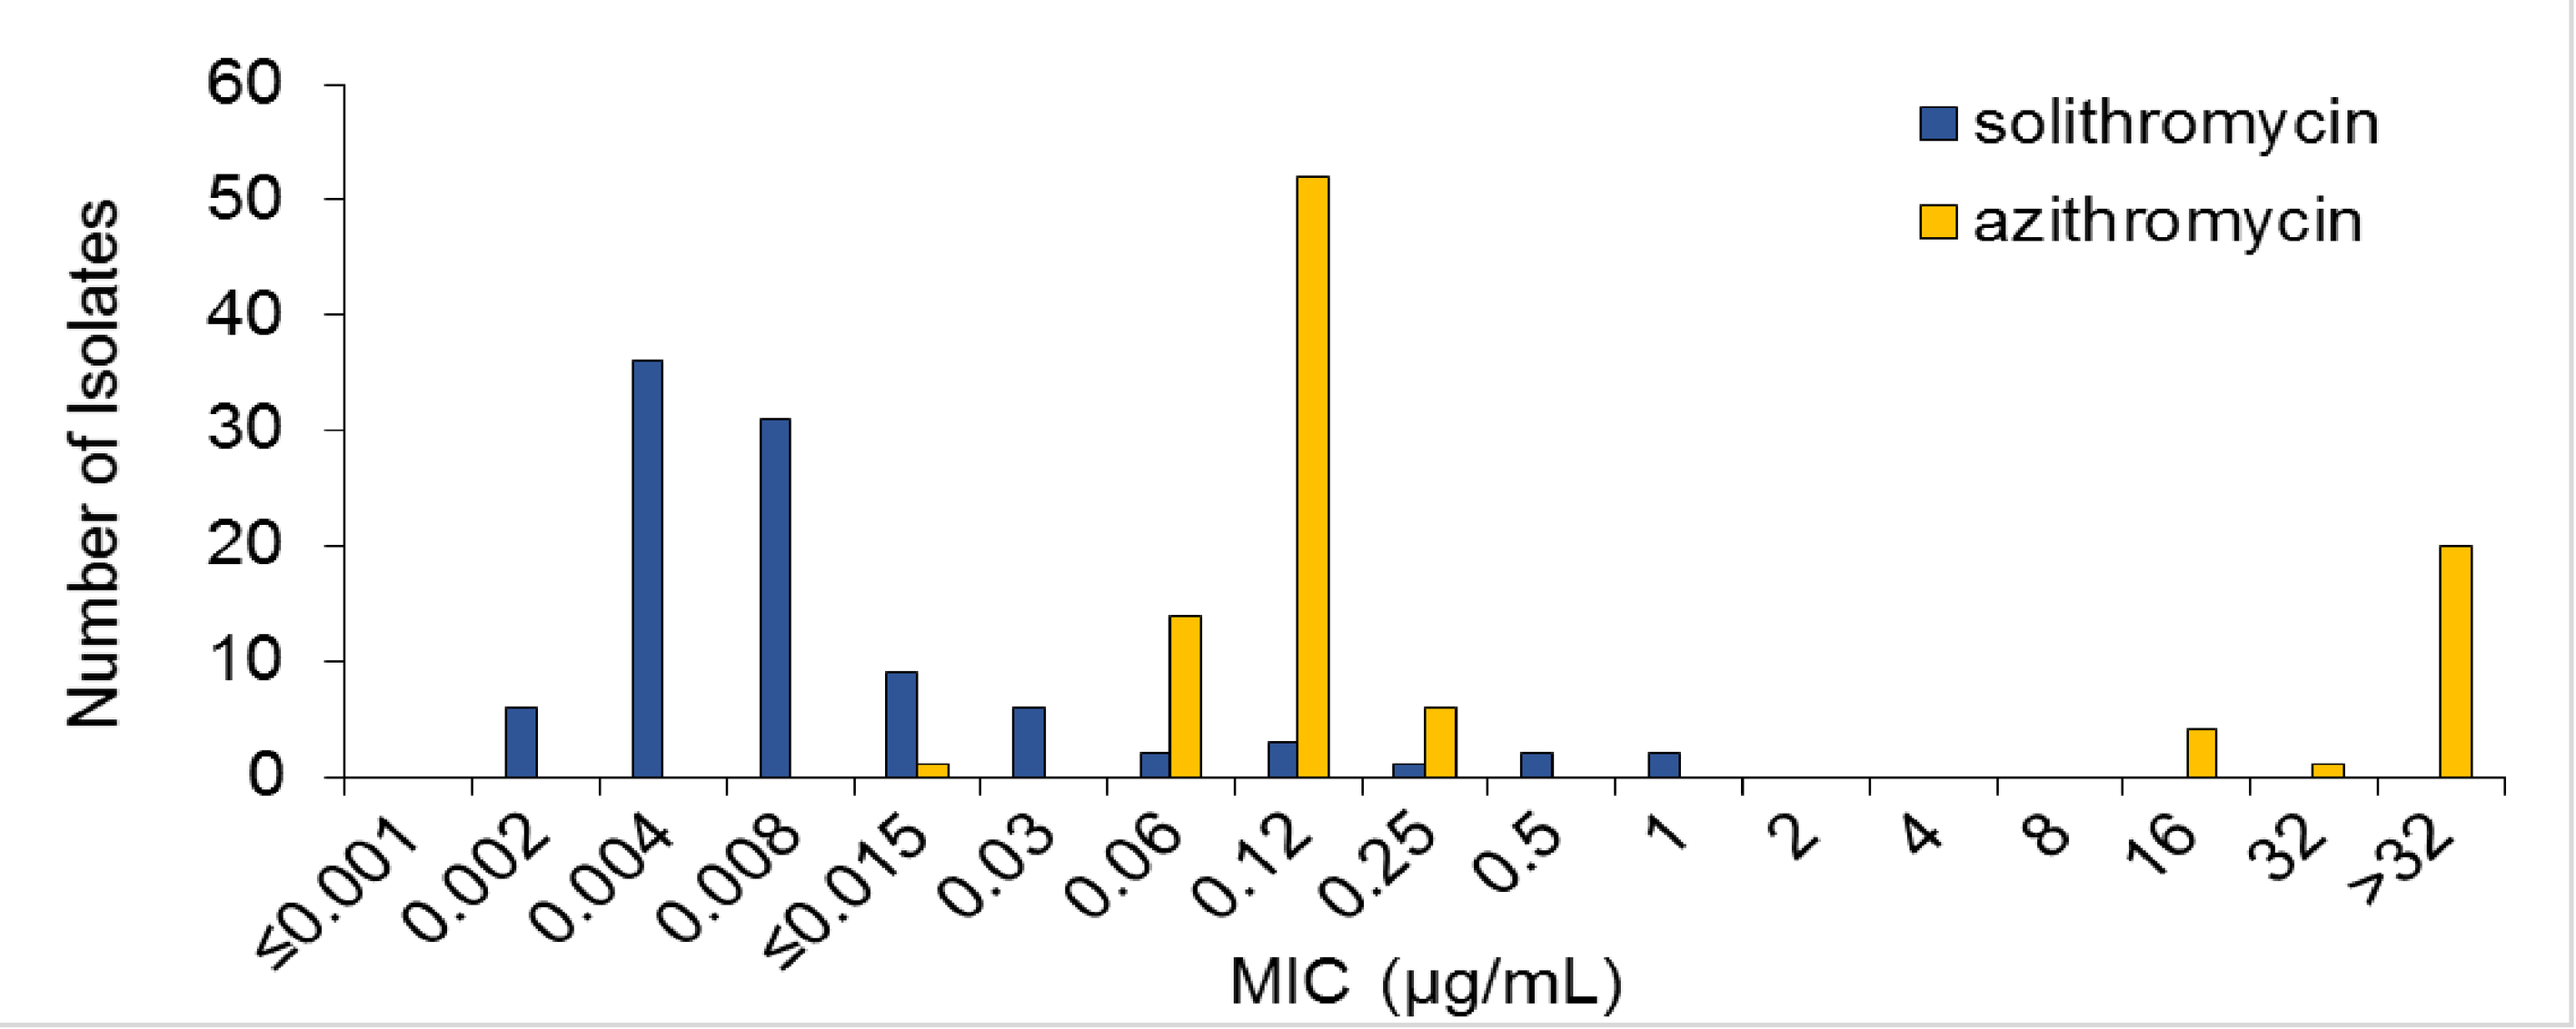
solithromycin
azithromycin
Number of Isolates
0
10
20
30
40
50
60
≤0.001
0.002
0.004
0.008
≤0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
>32
MIC (μg/mL)

Figure 3. Solithromycin MIC distribution among *S. pneumoniae*

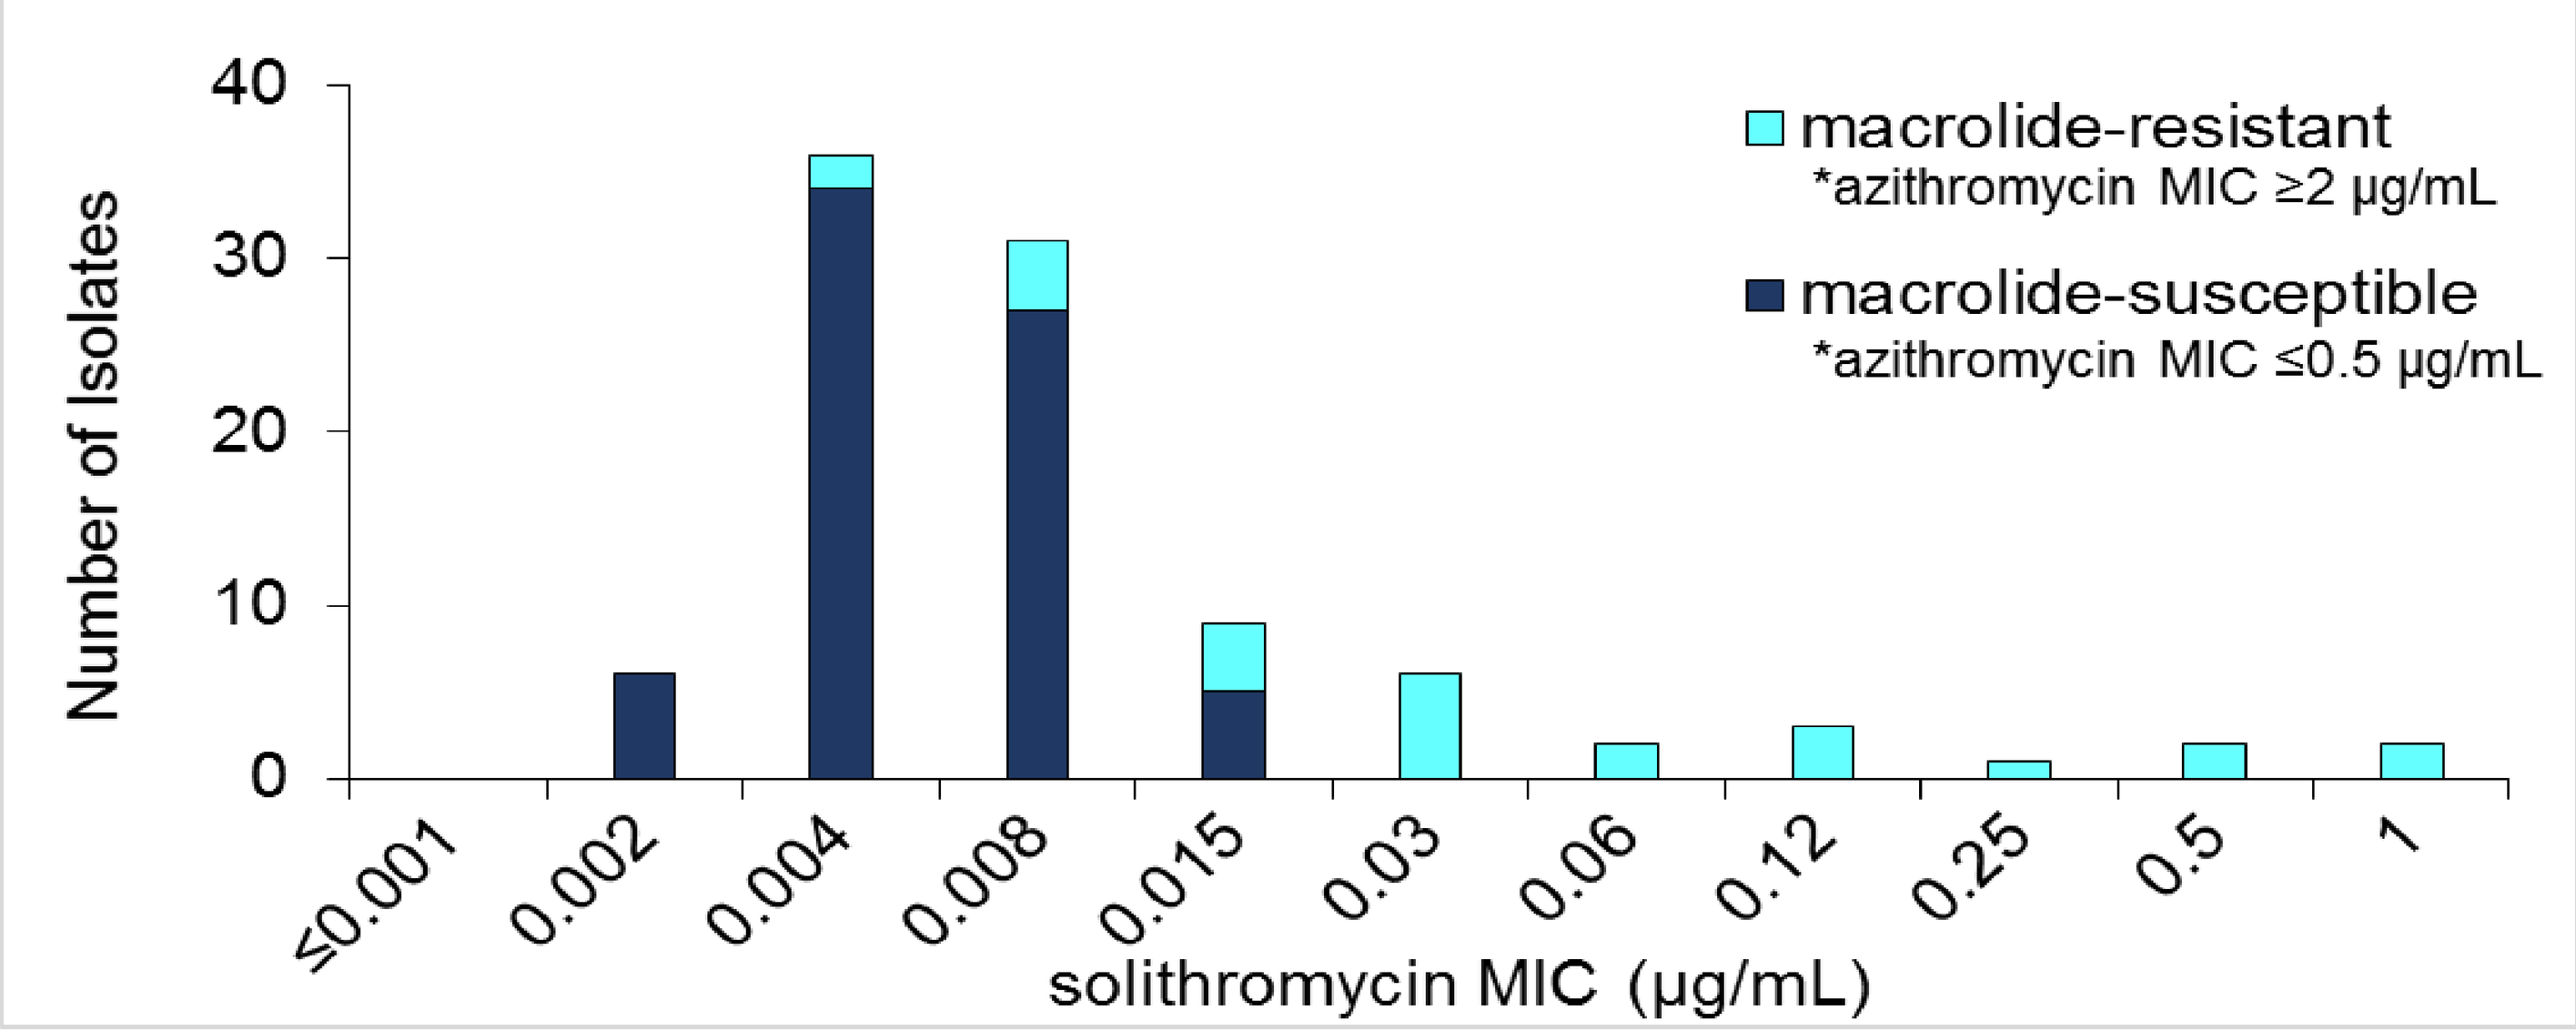
macrolide-resistant
*azithromycin MIC ≥2 μg/mL
macrolide-susceptible
*azithromycin MIC ≤0.5 μg/mL
Number of Isolates
0
10
20
30
40
≤0.001
0.002
0.004
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
solithromycin MIC (μg/mL)

- Most frequent pathogens: *S. pneumoniae* (18% of ITT), *M. pneumoniae* (8%), *H. influenzae, S. aureus, L. pneumophila* (all 4%).
- Majority of infections were monomicrobial (84%); Gram-positive + atypical diagnoses comprised majority (40%) of coinfections.
- Culture-based methods yielded diagnoses in 192 patients; use of molecular diagnostics enhanced pathogen detection by 70%.
- The rate of macrolide resistance in *S. pneumoniae* was 27%; SOLI maintained activity against these isolates (MICs ≤1 μg/mL).
- Among 37 *S. aureus* pneumonias, 92% were MSSA. One SOLI patient with HA-MRSA (MIC >32 μg/mL) was a failure at SFU.
- There was no correlation between clinical outcome and study drug MIC for key target pathogens shown in Table 3, suggesting target exposures were achieved.
- Relatively high rates of clinical success observed with SOLI against *Klebsiella pneumoniae* and *Pseudomonas aeruginosa* that were mostly comparable to or slightly better than those observed with MOXI are unexpected based on solithromycin MICs.

Table 2. Solithromycin MIC distribution for macrolide-resistant *S. pneumoniae* by genotype

Mac-R gene detected	0.004	0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	Total
ermB	2	3	4	4	2				2	17
mefA				1						1
mefE				1		3				4
ermB & mefA							1			1
ermB & mefE								2		2
negative		1								1

Table 3. By-pathogen Treatment Outcomes: Clinical Success at SFU and Outcome by MIC for selected CABP pathogens (ME-SFU population)

	Gram-positive						Gram-negative								Atypical			
	*S. pneumoniae**		β-haemolytic Strep		*S. aureus**		*H. influenzae**		*M. catarrhalis**		*K. pneumoniae*		*P. aeruginosa*		*M. pneumoniae**		*L. pneumophila**	
	SOLI	MOXI	SOLI	MOXI	SOLI	MOXI	SOLI	MOXI	SOLI	MOXI	SOLI	MOXI	SOLI	MOXI	SOLI	MOXI	SOLI	MOXI
By-pathogen Response	**62/74 (83.8)**	**62/69 (89.9)**	**2/2 (100.0)**	**1/1 (100.0)**	**17/20 (85.0)**	**16/16 (100.0)**	**15/16 (93.8)**	**19/20 (95.0)**	**4/4 (100.0)**	**3/3 (100.0)**	**5/7 (71.4)**	**2/3 (66.7)**	**4/4 (100.0)**	**5/6 (83.3)**	**29/34 (85.3)**	**25/27 (92.6)**	**17/18 (94.4)**	**15/16 (93.8)**
Outcome by MIC																		
≤0.000032															11/13 (84.6)		-	-
≤0.001																	-	-
0.002	2/3 (66.7)																-	-
0.004	14/17 (82.4)																-	-
0.008	12/15 (80.0)		1/1 (100.0)														-	-
0.015	4/4 (100.0)		1/1 (100.0)			3/3 (100.0)		9/9 (100.0)									-	-
0.03	3/3 (100.0)	1/1 (100.0)			1/2 (50.0)	5/5 (100.0)		3/4 (75.0)									-	-
0.06		9/10 (90.0)			13/13 (100.0)	4/4 (100.0)		4/4 (100.0)	3/3 (100.0)	3/3 (100.0)		1/1 (100.0)				1/1 (100.0)	-	-
0.12	2/2 (100.0)	31/35 (88.6)			3/4 (75.0)	1/1 (100.0)						1/2 (50.0)				11/11 (100.0)	-	-
0.25		1/1(100.0)		1/1 (100.0)					1/1 (100.0)								-	-
0.5	1/1 (100.0)																-	-
1							2/2 (100.0)							0/1 (0.0)			-	-
2						3/3 (100.0)	13/14 (92.9)							1/1 (100.0)			-	-
4														1/1 (100.0)			-	-
8								1/1 (100.0)									-	-
16											1/1 (100.0)			1/1 (100.0)			-	-
32 (or >16)											2/3 (66.7)			2/2 (100.0)			-	-
>32					0/1 (0.0)						2/3 (66.7)		4/4 (100.0)				-	-

* key target pathogens

Conclusions

Solithromycin is a new macrolide with potent activity against typical and atypical CABP pathogens, including macrolide-resistant isolates, and shows promise as a new macrolide for inpatient and outpatient treatment of CABP.
Microbiological diagnoses were enhanced, particularly for *S. pneumoniae* and *M. pneumoniae*, by the use of molecular techniques in addition to traditional culture-based methods, resulting in a diagnosis rate of 38%.
With overall and by-pathogen success rates similar to those observed in the SOLITAIRE-ORAL study, intravenous/IV-to-oral and oral solithromycin regimens were found to be non-inferior to moxifloxacin for treatment of CABP.

References

1. Mathers C, Ma Fat D, Boerma JT. World Health Organization. The Global Burden of Disease: 2004 Update. Geneva: World Health Organization; 2008. **2**. Donowitz GR. Acute pneumonia. In Mandell LA, Bennett JE, Dolin R, eds. Principles and Practice of Infectious Diseases. 7th ed. Philadelphia: Churchill Livingstone Elsevier; 2010; 891-916. **3**. Li HY, Guo Q, Song WD, Zhou YP, Li M, Chen XK, et al. Mortality among severe community-acquired pneumonia patients depends on combinations of 2007 IDSA/ATS minor criteria. Int J Infect Dis. 2015 Aug 6;38:141-145. **4**. File TM. *Streptococcus pneumoniae* and community-acquired pneumonia: a cause for concern. Am J Med. 2004; 117(3A):39S-50S. **5**. Niederman MS. Review of treatment guidelines for community-acquired pneumonia. Am J Med. 2004; 117(3A):51S-57S. **6**. Barrera CM, Mykietiuk A, Metev H, Nitu MF, Karimjee N, Doreski PA, et al. Efficacy and safety of oral solithromycin versus oral moxifloxacin for treatment of community-acquired bacterial pneumonia: a global, double-blind, multicentre, randomised, active-controlled, non-inferiority trial (SOLITAIRE-ORAL). Lancet Infect Dis. 2016 Apr;16(4):421:430.