Proposed Mechanisms to Explain the Unusual Visual Disturbances Associated with Telithromycin

***D. Bertrand*[1], S. Bertrand[1], P. Fernandes[2], [1] HiQScreen Sàrl, Geneva, Switzerland, [2] Cempra Pharmaceuticals, Chapel Hill, NC*

HiQScreen Sàrl
15, rue de l'Athénée
1206 Geneva
Switzerland
+41 786964656

Abstract

Background: Reports of macrolides causing "blurred vision" prompted investigation of the possible mechanisms of action. A first clue about blurred vision came from the relatively high prevalence of such symptoms reported by subjects exposed to telithromycin compared to structurally related macrolides (clarithromycin or azithromycin). As macrolides do not readily cross the blood brain barrier, blurred vision must arise from disruption of function in the peripheral nervous system. Since cholinergic neurotransmission plays a determinant role in peripheral nervous transmission we evaluated the possible interaction of macrolides with neuronal nicotinic acetylcholine receptors (nAChR) and compared the effects of telithromycin, clarithromycin, azithromycin and CEM-101, a novel fluoroketolide.
Methods: Effects of macrolides at human neuronal nAChRs were assessed using electrophysiological recordings in receptors expressed in *Xenopus* oocytes.
Results: Exposure to low concentrations of telithromycin (2 μM) for 15 minutes caused pronounced inhibition at the α3β4 (90 ± 10%) and α7 (85 ± 3%) receptor). Inhibition was significantly less with the other macrolides tested. Clarithromycin and azithromycin respectively inhibited the α3β4 receptor by 40 ± 7% and 56 ⁊ 10%, and the α7 receptor by 49 ± 4% and 51 ± 16%. CEM-101 inhibited the α3β4 receptor by 61 ± 4% and, importantly, had no effect at the α7 receptor (<6 ± 3%). As the α3β4 and α7 receptors are the major constituents of ganglionic transmission, inhibition of their activity will impair or even suppress neurotransmission in peripheral ganglia.
Conclusions: Dysfunction of the ciliary ganglion is expected to cause a loss of control of pupillary constriction and ciliary muscle contraction. Both effects thereby may combine to produce a reduction in the depth of field and of accommodation and cause a loss of focusing. This should result in profound vision disturbance and blurred vision for objects in the near and intermediate vision.

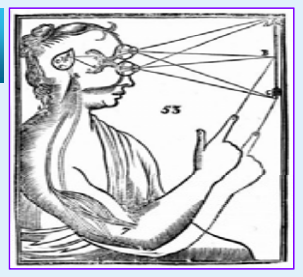

Introduction

Macrolides represent a large family of molecules displaying antibiotic activity that are used for the treatment of infections[1]. While macrolides such as erythromycin or clarithromycin are considered rather safe molecules, some unexplainable side effects were observed with telithromycin (Ketek) including aggravation of Myastenia Gravis, blurred vision, and liver toxicity. Myasthenia Gravis is preponderantly caused by the impairment the neurotransmission due to a dysfunction of the nicotinic acetylcholine receptors that act, through the chemical synapse, as a powerful amplification of the signal coming from the motor neuron nerve ending. Aggravation of the Myasthenia Gravis symptoms therefore suggests a possible interaction of macrolides with neuronal nicotinic acetylcholine receptors. As these receptors form a wide family of ligand gated channels that are expressed both in the central and peripheral nervous system where they control multiple critical body functions it is important to determine if and how macrolides could potentially interfere with this class of receptors. The aim of this work was to examine, using an electrophysiological approach, the effects of macrolides on the main nicotinic acetylcholine receptor subtypes.

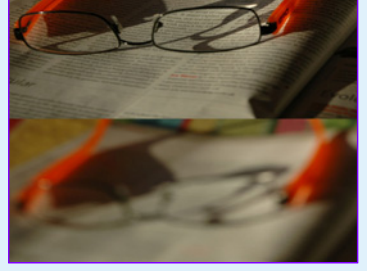

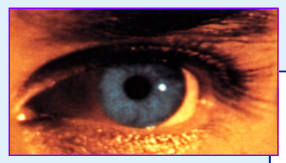

The eye

Or the brain

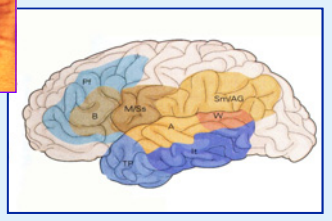

Methods

All experiments were carried out at human nAChRs expressed in *Xenopus* oocytes using the method of cDNA expression. *Xenopus* oocytes were prepared and injected using standard procedures. Briefly, ovaries were harvested from *Xenopus* Laevis females that have been deeply anesthetized and pitted following the animal rights rule from the Geneva canton. A small piece of ovary was isolated for immediate preparation while the remaining part was placed at 4 °C in a sterile Barth solution containing in mM NaCl 88, KCl 1, $NaHCO_3$ 2.4, HEPES 10, $MgSO_4.7H_2O$ 0.82, $Ca(NO_3)_2.4H_2O$ 0.33, $CaCl_2.6H_2O$ 0.41, at pH 7.4, and supplemented with 20 μg/ml of kanamycine, 100 unit/ml penicillin and 100 μg/ml streptomycin. All recordings were performed at 18 °C and cells superfused with OR2 medium containing in mM : NaCl 82.5, KCl 2.5, HEPES 5, $CaCl_2.2H_2O$ 1.8, $MgCl_2.6H_2O$ 1, pH 7.4, and 1 μM atropine was added to prevent possible activation of endogenous muscarinic receptors. Currents evoked by ACh or other agonists were recorded using an automated process equipped with standard two-electrode voltage-clamp configuration (TVEC). Unless indicated, cells were held at –70 mV. Data were captured and analyzed using a HiQScreen proprietary data acquisition and analysis software running under Matlab (Mathworks Inc.).

Results

Effects of macrolides were examined at four subtypes of nicotinic acetylcholine receptors (NMJ, α3β4, α4β2 and α7). While neuro muscular junction (NMJ) receptors are exclusively expressed on the muscle receptor membrane, the α3β4 receptors are preponderantly expressed in cholinergic ganglia. The α4β2 receptors represent the major high affinity nicotinic receptor in the brain (Figure 1). The homomeric α7 receptors are widely expressed in the central and peripheral nervous system and they have been shown to contribute to ganglionic neurotransmission [2-4].

cDNAs encoding for the human genes of these different subtype of receptors were injected into the nucleus of *Xenopus* oocytes and two days later properties of the receptors expressed at the cell membrane were investigated using two electrode recordings. Exposure of cells expressing functional nicotinic receptors to macrolides evoked no current indicating that these compounds do not act as agonists. A reduction of the responses evoked by ACh was, however, observed upon addition to the solution of a low concentration of macrolides (2 μM). The inhibition was dose dependent as shown in Figure 2. The amplitude of inhibition depends on both the macrolide and receptor subtype that confirms the specificity of inhibition.

To best understand macrolide effects, the relative activities of four macrolides (Clarithromycin, Azithromycin, Telithromycin and the novel CEM-101) were tested at the four nicotinic acetylcholine receptors. Experiments were carried out using sustained exposure to macrolides (20 minutes) and recording the acetylcholine evoked currents measured at periodic intervals. This protocol is thought to mimic the conditions experienced *in situ*. Results were quantified by plotting the fraction of acetylcholine-evoked current measured at the end of the twenty minutes incubation versus the amplitude of current measured in control. Figure 3 summarizes these results for the four macrolides at the different nicotinic acetylcholine receptors. The lower amplitude of the bar corresponding to the strongest inhibition. These data revealed that exposure to 2 μM telithromycin caused more than 80 % inhibition of the acetylcholine-evoked currents at the α3β4 and α7 nicotinic acetylcholine receptors (Table 1). As both receptor subtypes are mediating signal transmission in the peripheral cholinergic nervous system this indicates that exposure to telithromycin will inhibit neurotransmission in these ganglia.

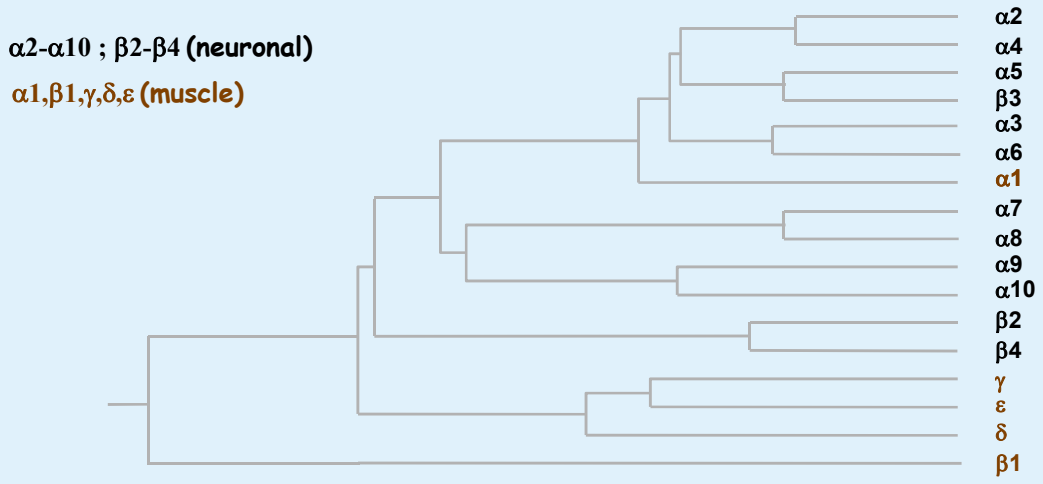
α2-α10 ; β2-β4 (neuronal)
α1,β1,γ,δ,ε (muscle)
α2
α4
α5
β3
α3
α6
α1
α7
α8
α9
α10
β2
β4
γ
ε
δ
β1

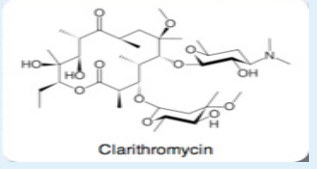
Clarithromycin

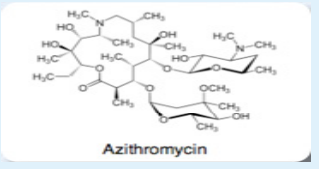
Azithromycin

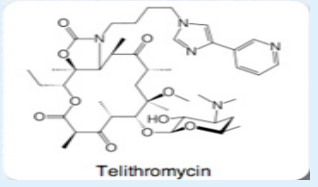
Telithromycin

Figure 1: Distribution of major nicotinic acetylcholine receptors

Effects of macrolides at nicotinic receptors

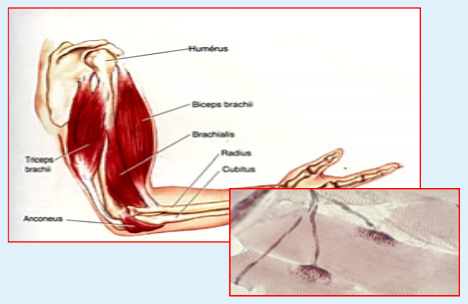
Humérus
Biceps brachii
Brachialis
Radius
Cubitus
Triceps brachii
Anconeus

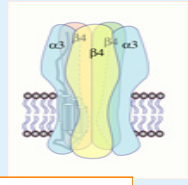

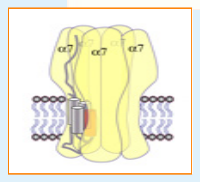

Figure 2

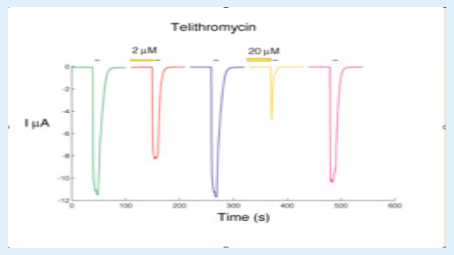
Telithromycin
2 μM
20 μM
I μA
Time (s)

Table 1

Compound (2μM)	α3β4	α7
Teli	90 ± 10%	85 ± 3%
Clari	40 ± 7%	49 ± 4%
Azi	56 ± 10%,	51 ± 16%.
CEM-101	61 ± 4%	<6 ± 3%

Figure 3

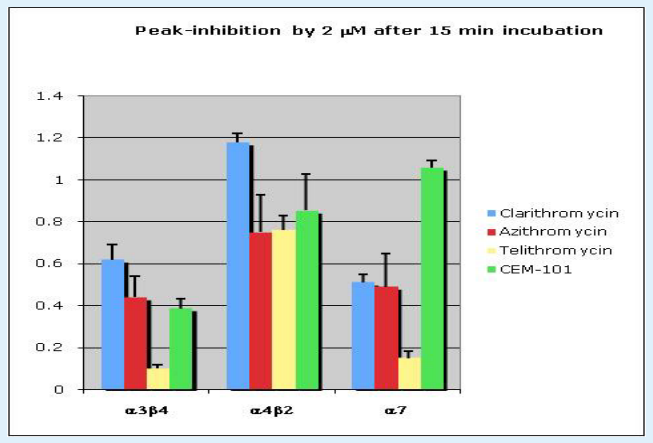
Peak-inhibition by 2 μM after 15 min incubation
1.4
1.2
1
0.8
0.6
0.4
0.2
0
α3β4
α4β2
α7
Clarithromycin
Azithromycin
Telithromycin
CEM-101

Conclusions

The observation that exposure to telithromycin aggravates two nicotinic acetylcholine receptor mediated effects, Myasthenia Gravis and visual function conditions suggested that macrolides may interact with the nicotinic acetylcholine receptors.

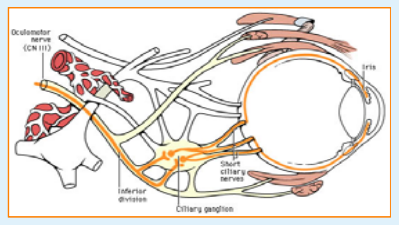
Oculomotor nerve (CN III)
Iris
Short ciliary nerves
Inferior division
Ciliary ganglion

Functional experiments carried out at human receptors revealed that macrolides can inhibit, in a dose and time dependent manner, these receptors function and that inhibition is receptor specific. The preferential inhibition observed with telithromycin at the ganglionic α3β4 and α7 receptor indicates that this molecule will impair ganglionic neurotransmission.
Proper functioning of these receptors is determinant for the neurotransmission in the ciliary ganglia and adequate command of muscle controlling the iris and crystalline lens accommodation. Inhibition of the α3β4 and α7 receptors is therefore expected to cause a dysfunction of the ciliary ganglia that can explain the blurred vision reported by patients taking telithromycin.

References

1. Omura *Macrolide Antibiotics Chemistry, Biology and Practice* 2002).
2. Dani, J. A. & Bertrand, D. Nicotinic acetylcholine receptors and nicotinic cholinergic mechanisms of the central nervous system. *Annu Rev Pharmacol Toxicol* **47,** 699-729 (2007).
3. Berg, D. K. & Conroy, W. G. Nicotinic alpha 7 receptors: synaptic options and downstream signaling in neurons. *J Neurobiol* **53,** 512-523 (2002).
4. Sargent, P. B. Nicotinic receptors concentrated in the subsynaptic membrane do not contribute significantly to synaptic currents at an embryonic synapse in the chicken ciliary ganglion. *J Neurosci* **29,** 3749-3759 (2009).