F1-2151

Development of an Intravenous Formulation of Solithromycin (CEM-101), a Novel Potent Fluoroketolide

P. Fernandes, T. Degenhardt, D. Pereira

Cempra Pharmaceuticals, Inc., Chapel Hill, North Carolina

2010 ICAAC, Boston, MA
Prabhavathi Fernandes, Ph.D..
6340 Quadrangle Drive, Suite 100
Chapel Hill, North Carolina 27517
prabha@fernandes-domain.com

Abstract

Background: Solithromycin (CEM-101) is a potent new fluoroketolide under development for the treatment of bacterial respiratory tract and other infections. In order to treat moderate to moderately severe to severe community acquired bacterial pneumonia (CABP), an intravenous formulation of Solithromycin is desirable.
Methods: A soluble, stable formulation of Solithromycin was developed and tested in 28-day toxicology studies in dogs and monkeys.
CEM-101 was infused once daily at doses of 0, 5, 10 and 15 mg/kg in dogs and at doses of 0, 5, 12.5 and 25 mg/kg in monkeys. Blood was drawn pre-dose and at 0, 0.5, 1, 4, 8, 12 (monkey) and 24 hours following cessation of dosing on days 1 and 28 for pharmacokinetic measurements. Clinical pathology and histopathology evaluations were performed.
Results: Unlike other macrolides, it was noted that there was no apparent pain and no significant irritation at the injection sites with Solithromycin. Excellent blood levels were achieved in both species. In the dog, the C_{max} of the 15 mg/kg dose group on days 1 and 28 were 3.9 mg/L and 3.0 mg/L, respectively. There were no toxicologically significant serum chemistry, hematology or coagulation changes. Injection site had minor microscopic changes, mostly procedure related. Monkey plasma C_{max} at 25 mg/kg were 5.4 mg/L on Day 1 and 5.9 mg/L on Day 28. There were no toxicologically significant serum chemistry, hematology or coagulation changes. Injection sites had minor microscopic changes, mostly procedure related. Overall, there was little variability in blood levels and no cardiac or other pharmacological abnormalities were noted in the dog or monkey.
Conclusion: The toxicology assessment for the intravenous product of Solithromycin has been successfully completed. Solithromycin was well tolerated with only minimal and reversible findings. Solithromycin is the first macrolide/ketolide since azithromycin to have the potential for an intravenous formulation making it feasible to conduct trials with a macrolide in moderately severe to severe CABP.

Background

The oral formulation of Solithromycin is currently in Phase 2 clinical trials in moderate to moderately-severe community acquired bacterial pneumonia (CABP).

Solithromycin has potent in vitro activity against the key bacterial pathogens associated with CABP, including macrolide-resistant strains and atypical bacteria.

Solithromycin is well distributed in the blood, organs, tissues and cells in nonclinical studies, and penetrates extensively into human lung ELF and AM. Taste was tolerable. It is not highly bound to plasma proteins.

Single-dose and multiple-dose human PK support a once-daily oral dosing regimen. The oral bioavailability of Solithromycin is not influenced by food.

Solithromycin has an expanded spectrum of activity that will allow evaluation in areas of unmet medical need, including gonococcal and nongonococcal urethritis/cervicitis, malaria, tuberculosis, Hansen's disease, *Mycobacterium avium*-intracellulare complex (MAC), and biodefense pathogens (*Bacillus anthracis* and *Francisella tularensis*).

Solithromycin has anti-inflammatory properties that may enhance clinical efficacy in infections.

Solithromycin has in vitro intracellular activity, and shows efficacy against intracellular (intraphagocytic) pathogens in vivo.

Solithromycin has no significant inhibitory effect on the nicotinic acetylcholine receptor subtype, in contrast to telithromycin. Some of the adverse effects observed with telithromycin appear to be associated with the inhibition of nicotinic acid acetylcholine receptor subtypes.

Unlike older macrolides, solithromycin did not activate the motilin receptor in vitro, and as a result, it is not expected to induce gastrointestinal contractions through the motilin receptor response.

Solithromycin (CEM-101) is the first fluoroketolide in development

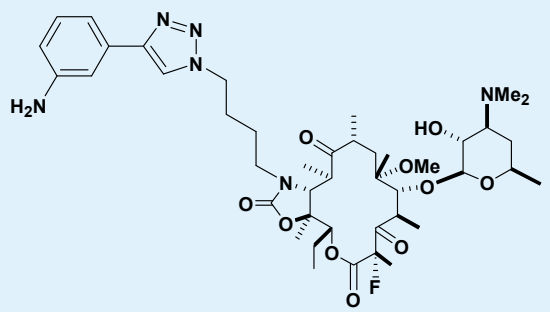

N
N
N
H$_2$N
NMe$_2$
HO
OMe
O
O
O
N
O
O
O
O
O
F
O

Introduction

An intravenous (IV) formulation of solithromycin is being developed in an effort to provide treatment for sicker patients with more severe CABP. The IV formulation followed by the oral formulation could allow step-down therapy for appropriate patients, and thereby provide a benefit over an oral formulation alone

Many experiments have been conducted toward the development of a suitable IV formulation, and were based upon past experience and available information on IV formulations of macrolides.

Materials and Methods

Solithromycin IV formulation is a buffered isotonic solution.

Animals:

Beagle dogs (Marshall BioResources, Inc, North Rose, NY), 6 to 7 months old, body weights ranged from 7.1 to 9.4 kg for males and from 5.3 to 6.8 kg for females.

Cynomolgus monkeys (Worldwide Primates, Inc., Miami, FL), 3 to 4 years old, body weights ranged from 2.4 to 4.6 kg for males and 2.2 to 3.5 kg for females.

28-day Repeat-dose Toxicity in Dogs
Solithromcyin was administered to beagle dogs at doses of 0, 5, 10 and 15 mg/kg via intravenous infusion into the saphenous or cephalic veins for 28 days.

28-day Repeat-dose Toxicity in Monkeys
Solithromcyin was administered to cynomolgus monkeys at doses of 0, 5, 12.5 and 25 mg/kg via intravenous infusion into the saphenous or brachial veins for 28 days.

Discussion

CEM-101 was well tolerated in dogs and monkeys after repeated daily IV dosing for 28 days as infusions into saphenous, cephalic, or brachial veins.

Solithromycin is expected to be the first ketolide available in both IV and oral formulations. The IV formulation will allow for treatment of patients requiring parenteral therapy, either due to the severity of their infection, inability to tolerate oral medications or presence of gastrointestinal disorders leading to malabsorption, with the option of oral step-down in appropriate patients.

Unlike older macrolides, the apparent lack of pain or discomfort upon injection, in both dogs and monkeys, could be a benefit of this IV antibiotic formulation.

Solithromycin IV formulation was developed and an IND for the intravenous formulation was filed. The Intravenous Phase 1 study is being initiated in Q4, 2010.

Results

28-day Repeat-dose Toxicity in Dogs

Solithromycin at doses up to 10 mg/kg/d by intravenous infusion for 28 consecutive days was well tolerated. Administration of 15 mg/kg/d resulted in the premature euthanasia of one male dog but was well tolerated in the remaining dogs. The cause of death in the one dog was unknown but the animals did have reduced fecal output and decreased body weight. A slight to moderate treatment-related decrease in mean body weight, associated with decreased food consumption, was noted in the 15 mg/kg/d animals of both sexes on Day 28; this was fully reversible.

The majority of the infusion site lesions were also seen in the control animals at lower incidence and/or severity, suggesting that the changes were predominantly procedure-related, but slightly exacerbated by the administration of solithromycin. All of the changes were completely or partially reversed by the end of the 28-day recovery period. There were no changes in ophthalmology, electrocardiography or coagulation that could be attributed to the administration of solithromycin.

A slight decrease in urine volume noted on Day 29 in the animals of both sexes at 15 mg/kg/d was also completely reversed at the end of the 28-day recovery period.

Liver Enzymes: Marginal increases in mean ALT and cholesterol levels, slightly above the historical normal ranges, were noted on Day 29 in the animals of both sexes at all dose levels. In view of the small magnitude of the changes and in the absence of any histopathological correlates, the toxicological significance of these changes remains equivocal. At the end of the 28-day recovery period, all values were comparable to concurrent controls.

Plasma Pharmacokinetics: Solithromycin exposures (C_{max} and $AUC_{(0-24)}$) increased with increasing solithromycin dose in both sexes on Days 1 and 28. The plasma exposure of solithromycin was similar in both males and females on Day 1 and slightly higher in males on Day 28. Solithromycin appeared to be eliminated more slowly at high doses. The blood levels are shown in Figure 1.

Figure 1

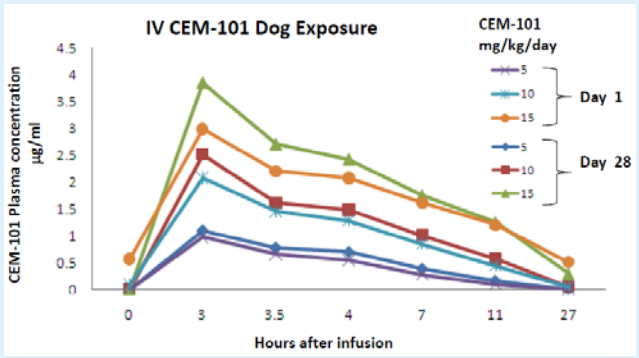

IV CEM-101 Dog Exposure
CEM-101 mg/kg/day
5
10
15
Day 1
5
10
15
Day 28
CEM-101 Plasma concentration µg/ml
4.5
4
3.5
3
2.5
2
1.5
1
0.5
0
0
3
3.5
4
7
11
27
Hours after infusion

28-day Repeat-dose Toxicity in Monkeys

Solithromycin at doses up to 25 mg/kg/d for 28 consecutive days was well tolerated and resulted mainly in local lesions at the infusion sites. The majority of the infusion site lesions were also seen in the control animals at lower incidence and/or severity, suggesting that the changes were predominantly procedure-related, but slightly exacerbated by the administration of solithromycin.

All of the solithromycin related changes were completely reversed by the end of the 28-day recovery period.

There were no changes in body weights, ophthalmology, electrocardiography, hematology, coagulation, urinalysis or organ weights that could be attributed to the administration of solithromycin by intravenous infusion at doses up to 25 mg/kg/d for 28 consecutive days.

Liver enzymes: Marginal increases in mean ALT and AST levels were noted on Day 29 in the 12.5 and 25 mg/kg/d males. These changes were not considered to be toxicologically significant, as most values remained within the normal variation range for this species or were slightly above the historical normal ranges for cynomolgus monkeys but not statistically different from controls and had no histopathological correlates. At the end of the 28-day recovery period (Day 57), all values were comparable to concurrent controls.

Pharmacokinetics: Dose dependent increases in plasma levels were noted. The plasma levels on day 1 and day 28 in each of the dose groups are shown in Figure 2.

In the dog and monkey, plasma concentrations of the primary metabolites, N-acetyl solithromycin and CEM-214, were very low.

Figure 2

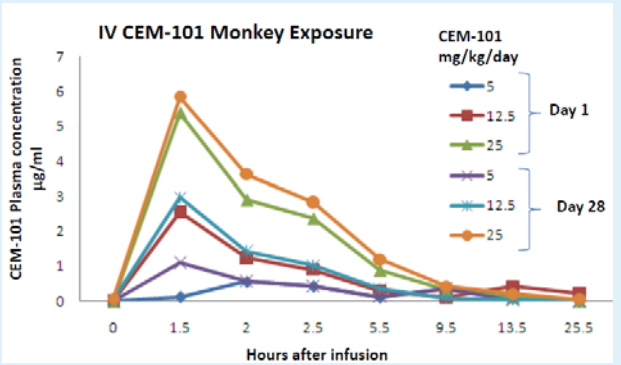

IV CEM-101 Monkey Exposure
CEM-101 mg/kg/day
5
12.5
25
Day 1
5
12.5
25
Day 28
CEM-101 Plasma concentration µg/ml
7
6
5
4
3
2
1
0
0
1.5
2
2.5
5.5
9.5
13.5
25.5
Hours after infusion

References

1. Farrell DJ, Sader HS, Castanheira M, Biedenbach DJ, Rhomberg PR, Jones RN. Antimicrobial characterisation of CEM-101 activity against respiratory tract pathogens, including multidrug-resistant pneumococcal serogroup 19A isolates. Int J Antimicrob Agents. 35: 537-543. 2010.
2. Roblin PM, Kohlhoff SA, Parker C, Hammerschlag MR. In vitro activity of CEM-101, a new fluoroketolide antibiotic, against *Chlamydia trachomatis* and *Chlamydia (Chlamydophila) pneumoniae*. Antimicrob Agents Chemother. 54:1358-1359. 2010.
3. McGhee P, Clark C, Kosowska-Shick KM, Nagai K, Dewasse B, Beachel L, Appelbaum PC. In vitro activity of CEM-101 against *Streptococcus pneumoniae* and *Streptococcus pyogenes* with defined macrolide resistance mechanisms. Antimicrob Agents Chemother. 54:230-238. 2010.
4. Lemaire S, Van Bambeke F, Tulkens PM. Cellular accumulation and pharmacodynamic evaluation of the intracellular activity of CEM-101, a novel fluoroketolide, against *Staphylococcus aureus*, *Listeria monocytogenes*, and *Legionella pneumophila* in human THP-1 macrophages. Antimicrob Agents Chemother. 53:3734-3743. 2009.
5. Waites KB, Crabb DM, Duffy LB. Comparative in vitro susceptibilities of human mycoplasmas and ureaplasmas to a new investigational ketolide, CEM-101. Antimicrob Agents Chemother. 53:2139-2141. 2009.
6. Bertrand, D., Bertrand, S., Neveu, E., Fernandes, P. Molecular Characterization of Off-Target Activities of Telithromycin, a Macrolide/Ketolide Antibiotic: The Role of Nicotinic Acetylcholine Receptors. 2010. [abstract # 252]. Infectious Diseases Society of America Annual Meeting (Vancouver Canada). Vancouver Canada: Infectious Diseases Society of America, 2010.
7. Still, J.G., Clark, K., Degenhardt, T., Scott, D., Fernandes, P., Gutierrez, M.J. Single Oral Dose Pharmacokinetics and Safety of CEM-101 in Healthy Subjects. 2008. [abstract # F1-3972a]. 48th Interscience Conference on Antimicrobial Agents and Chemotherapy (Washington DC). Washington DC: American Society for Microbiology, 2008.
8. Rodvold, K.A., Gotfried, M.H., Okusanya, O.O., Forrest, A., Bhavnani, S.M., Still, J.G., Clark, K., Fernandes, P., Ambrose, P.G. Intrapulmonary Penetration of CEM-101 In Healthy Adult Subjects. 2010. [abstract # A1-690]. 50th Interscience Conference on Antimicrobial Agents and Chemotherapy (Boston MA). Boston MA: American Society for Microbiology 2010.